Allogenic CAR T-cells Targeting CD123 for Adoptive Immunotherapy of Acute Myeloid Leukemia (AML)

Roman Galetto[1], Céline Lebuhotel[1], Patricia Françon[1], Agnès Gouble[1] and Julianne Smith[1]

[1] Cellectis SA, Paris, France

#1 Abstract

Chimeric antigen receptor (CAR)-redirected T-cells have given rise to long-term durable remissions and remarkable objective response rates in patients with refractory leukemia, raising hopes that a wider application of CAR technology may lead to a new paradigm in cancer treatment. A limitation of the current autologous approach is that CAR T-cells must be manufactured on a "per patient basis". We have developed a standardized platform for manufacturing T-cells from third-party healthy donors to generate allogeneic "off-the-shelf" engineered CAR+ T-cell–based frozen products. This platform utilizes Transcription Activator-Like Effector Nuclease (TALEN) gene editing technology to inactivate the TCRα constant (TRAC) gene, eliminating the potential for T-cells bearing alloreactive TCR's to mediate Graft versus Host Disease (GvHD). We have previously demonstrated that editing of the TRAC gene can be achieved at high frequencies, obtaining up to 80% of TCRα negative cells. This allows us to efficiently produce TCR-deficient T-cells that have been shown to no longer mediate alloreactivity in a xeno-GvHD mouse model. In addition to CAR expression, our T-cells are engineered to co-express the RQR8 gene as a safety feature, with the aim of rendering them sensitive to the monoclonal antibody rituximab.

In this work we present the adaptation of this allogeneic platform to the production of T cells targeting CD123, the transmembrane alpha chain of the interleukin-3 receptor, which is expressed in tumor cells from the majority of patients with Acute Myeloid Leukemia (AML). To identify an effective CAR targeting CD123, we have screened multiple antigen recognition domains in the context of several different CAR architectures to identify candidates displaying activity against cell lines expressing variable levels of the CD123 antigen. Furthermore, experiments in an AML mouse model using anti-CD123 CAR T cells demonstrate important anti-tumor activity in vivo. The ability to carry out large scale manufacturing of allogeneic, non alloreactive CD123 specific T cells from a single healthy donor will thus offer the possibility of an off-the-shelf treatment that would be immediately available for administration to a large number of AML patients.

#2 Allogenic approach targeting CD123 to treat AML

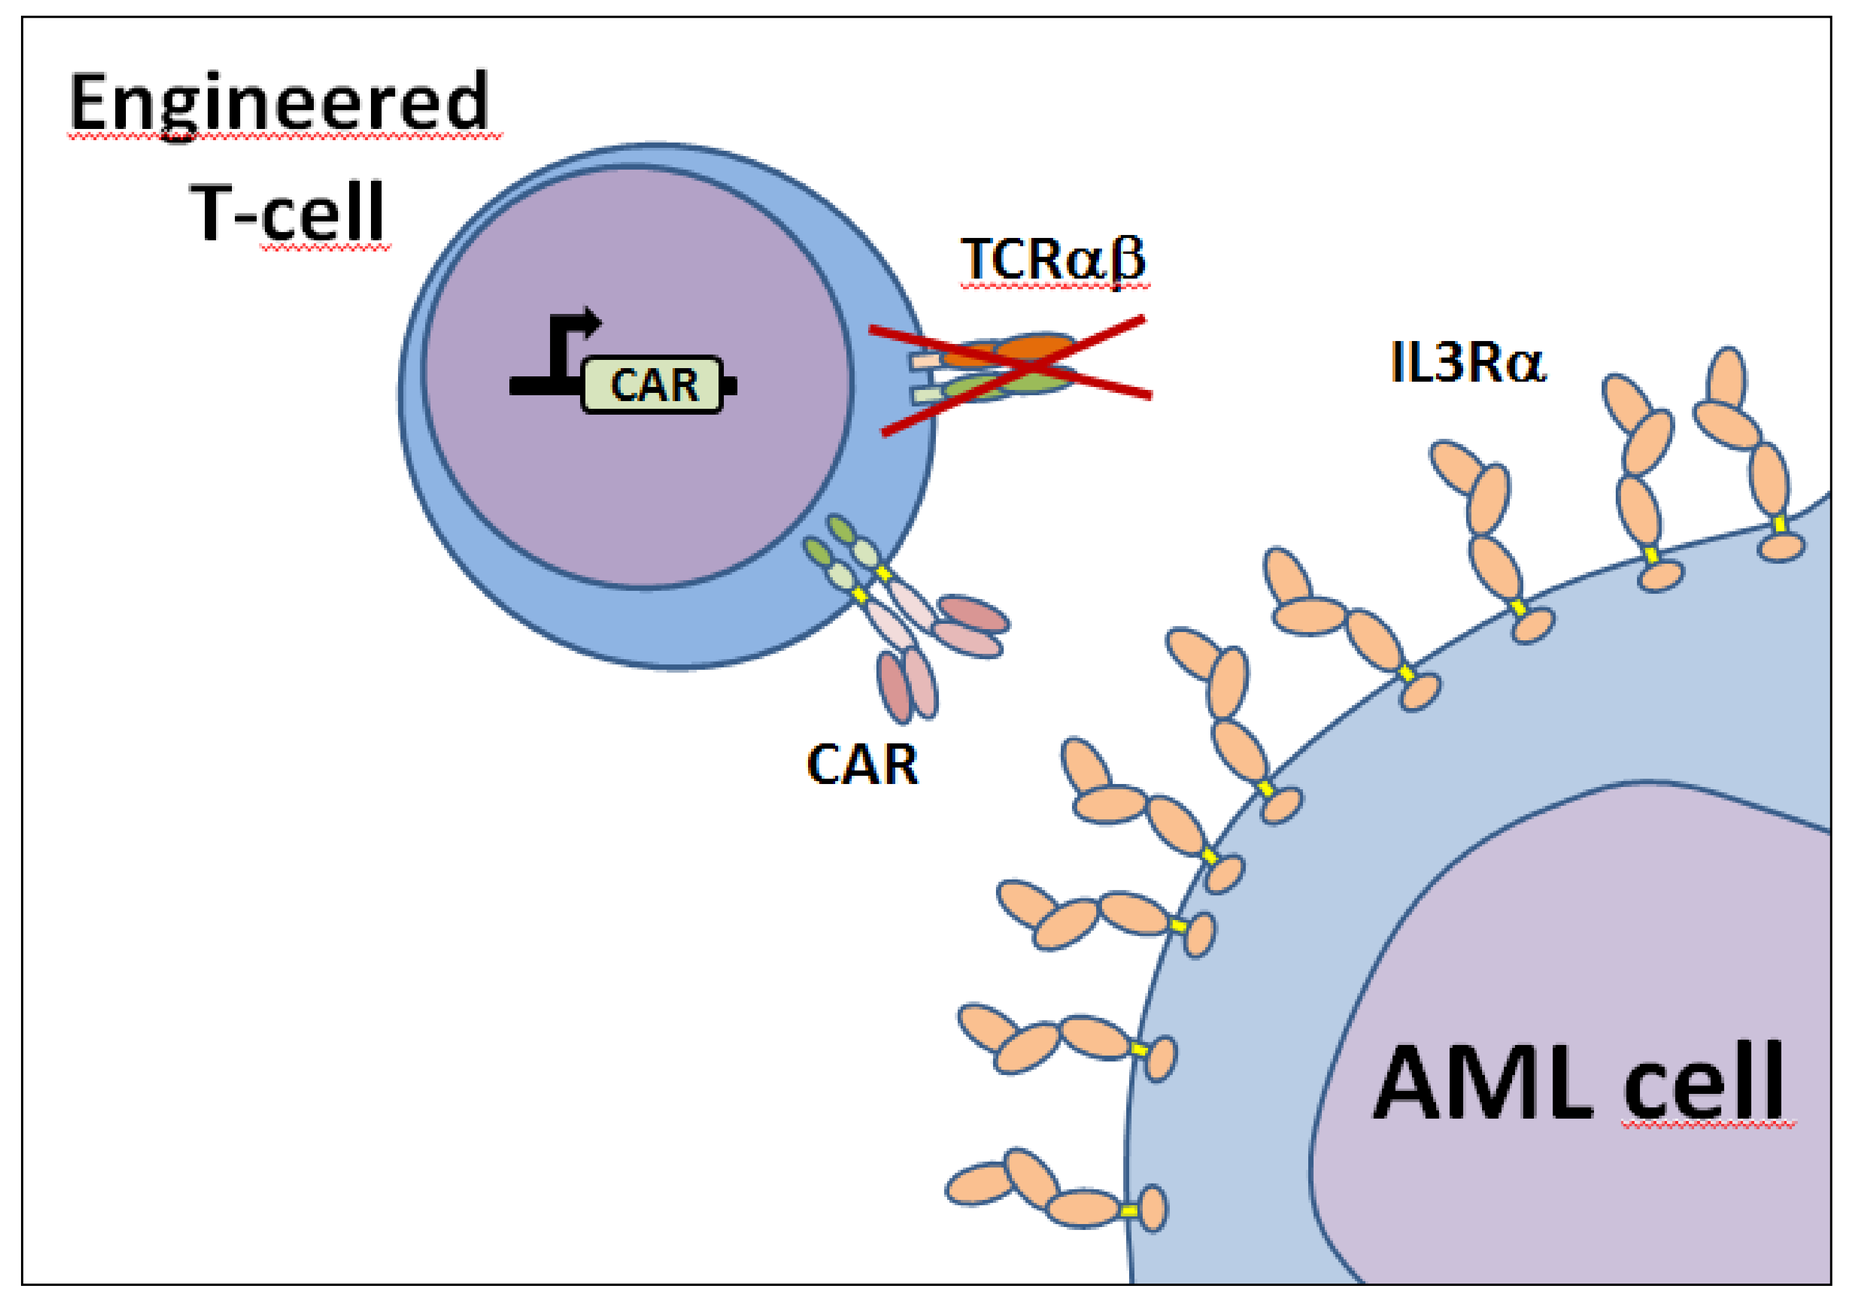

Engineered
T-cell
CAR
TCRαβ
IL3Rα
CAR
AML cell

One of the key barriers to the adoptive transfer of 3rd party CAR T-cells can be overcome via the application of TALEN gene editing technology, deletion of the TCR gene and thus the use of any donors' T-cells without the risk of GvHD.

#3 TCR alpha inactivation prevents Graft vs. Host disease

TALEN were designed to recognize and cleave the coding sequence of human TCRα gene

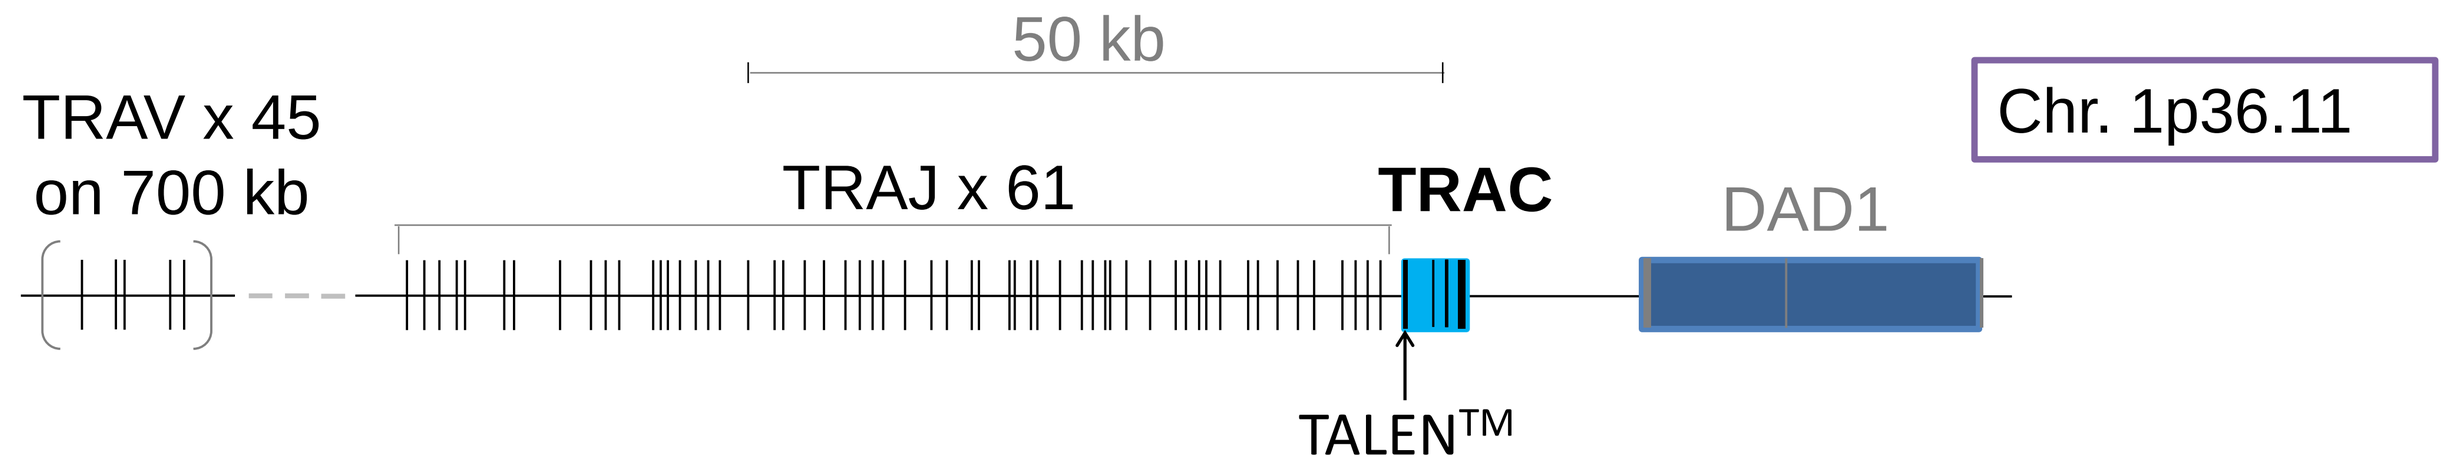

50 kb
Chr. 1p36.11
TRAV x 45
on 700 kb
TRAJ x 61
TRAC
DAD1
TALEN™

High levels of gene inactivation can be obtained in primary T cells by electrotransfer of TALEN mRNAs using a PulseAgile® electroporation technology.

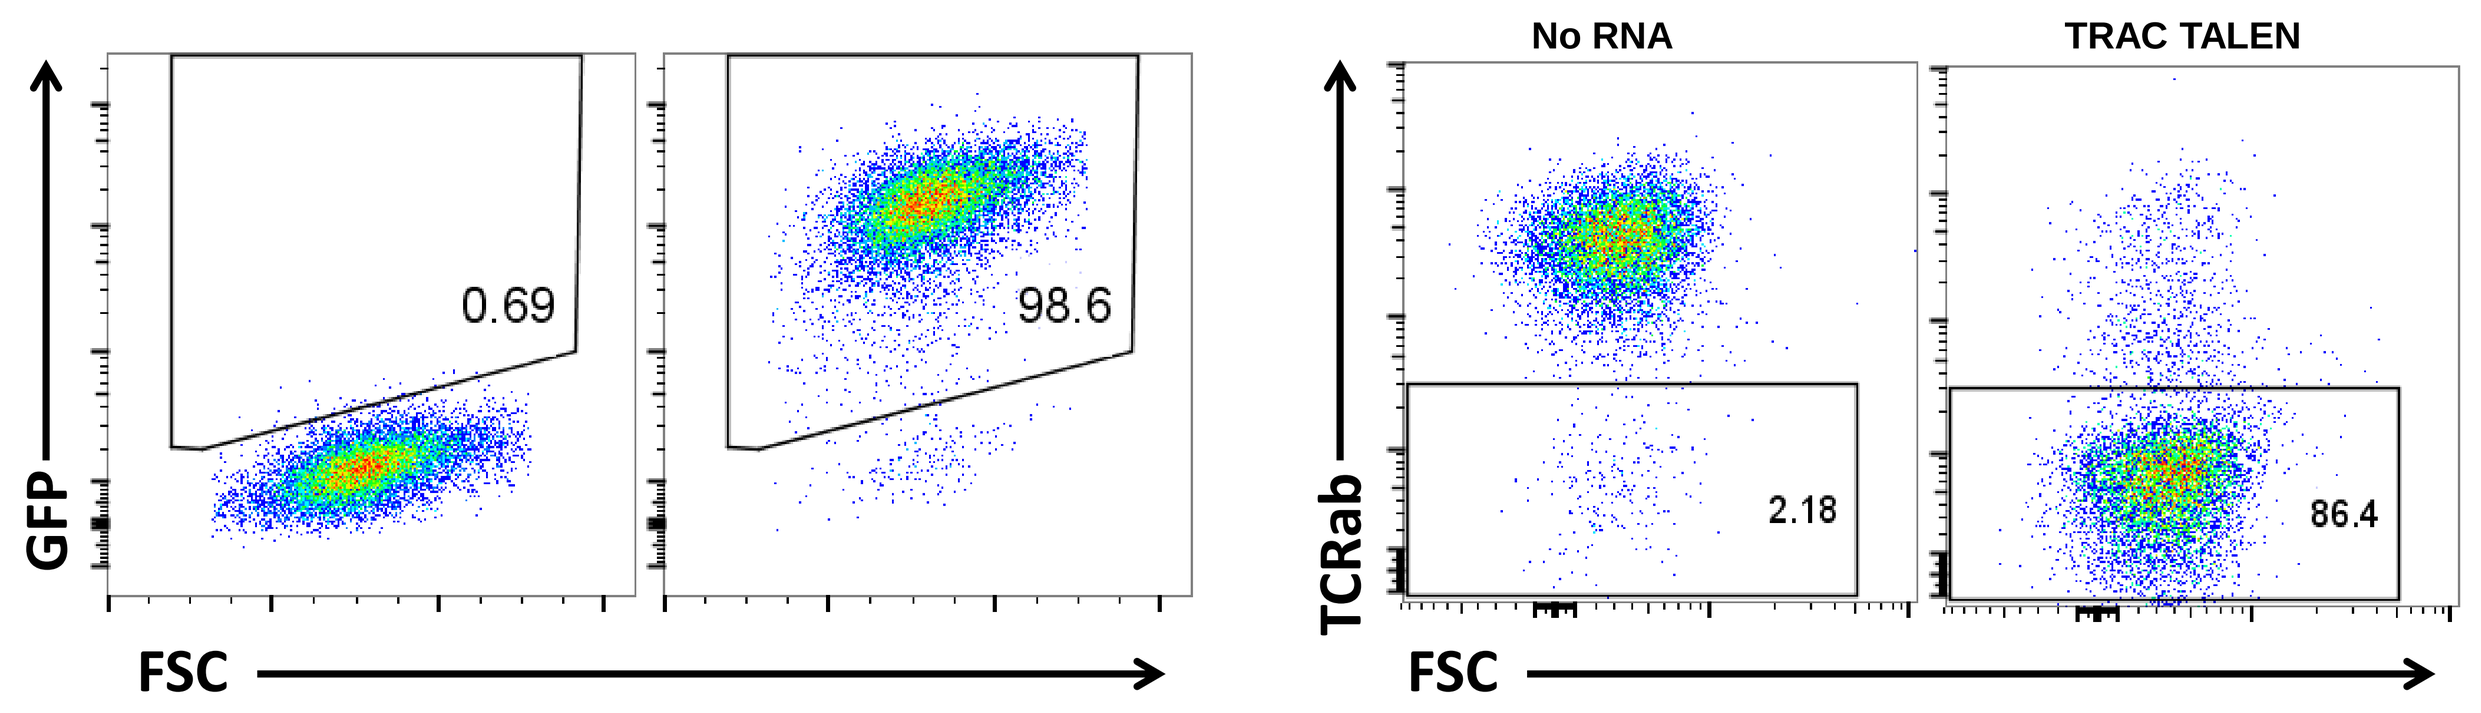

0.69
98.6
GFP
FSC
No RNA
TRAC TALEN
2.18
86.4
TCRab
FSC

Xenogeneic model of Graft vs. Host Disease: Irradiated NOG mice were injected with PBS (group 1), or 30x10^6 human T cells from the same donor. The cells were obtained from activated PBMCs after 3 day expansion (group 2), 15-day expansion (group 3), mock electroporation and 15-day expansion (group 4), or TRAC TALEN-electroporation, 15-day expansion and depletion of TCR+ cells (group 5).

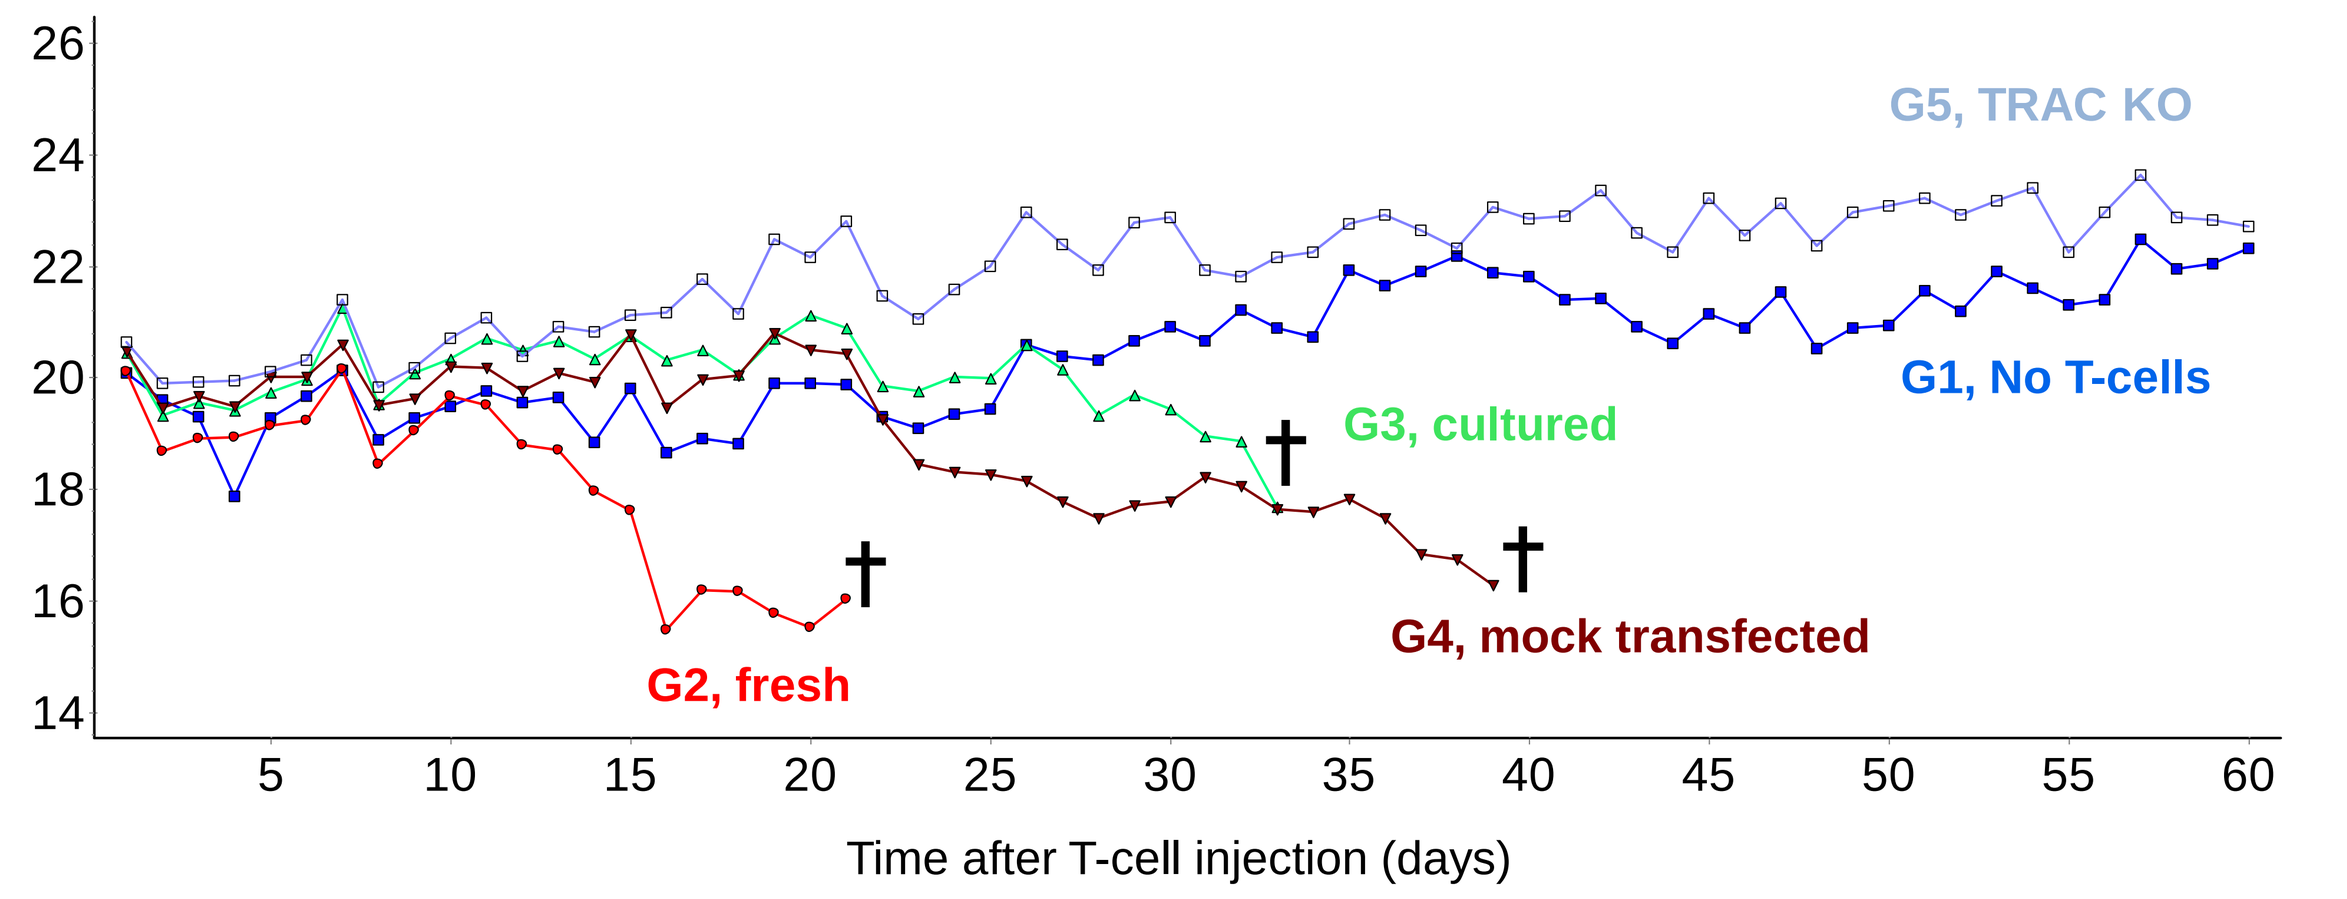

G5, TRAC KO
G1, No T-cells
G3, cultured
G2, fresh
G4, mock transfected
Time after T-cell injection (days)

No mice receiving TRAC TALEN-treated cells showed signs of GvHD, whereas all mice receiving non engineered cells developped GvHD and had to be sacrificed. Human cells survived at least 2 weeks in all groups.

#4 Generation of anti-CD123 Chimeric Antigen Receptors

An scFv targeting the extracellular domain of the human CD123 antigen was used to construct three chimeric antigen receptors, each containing hinge sequences of different lengths.

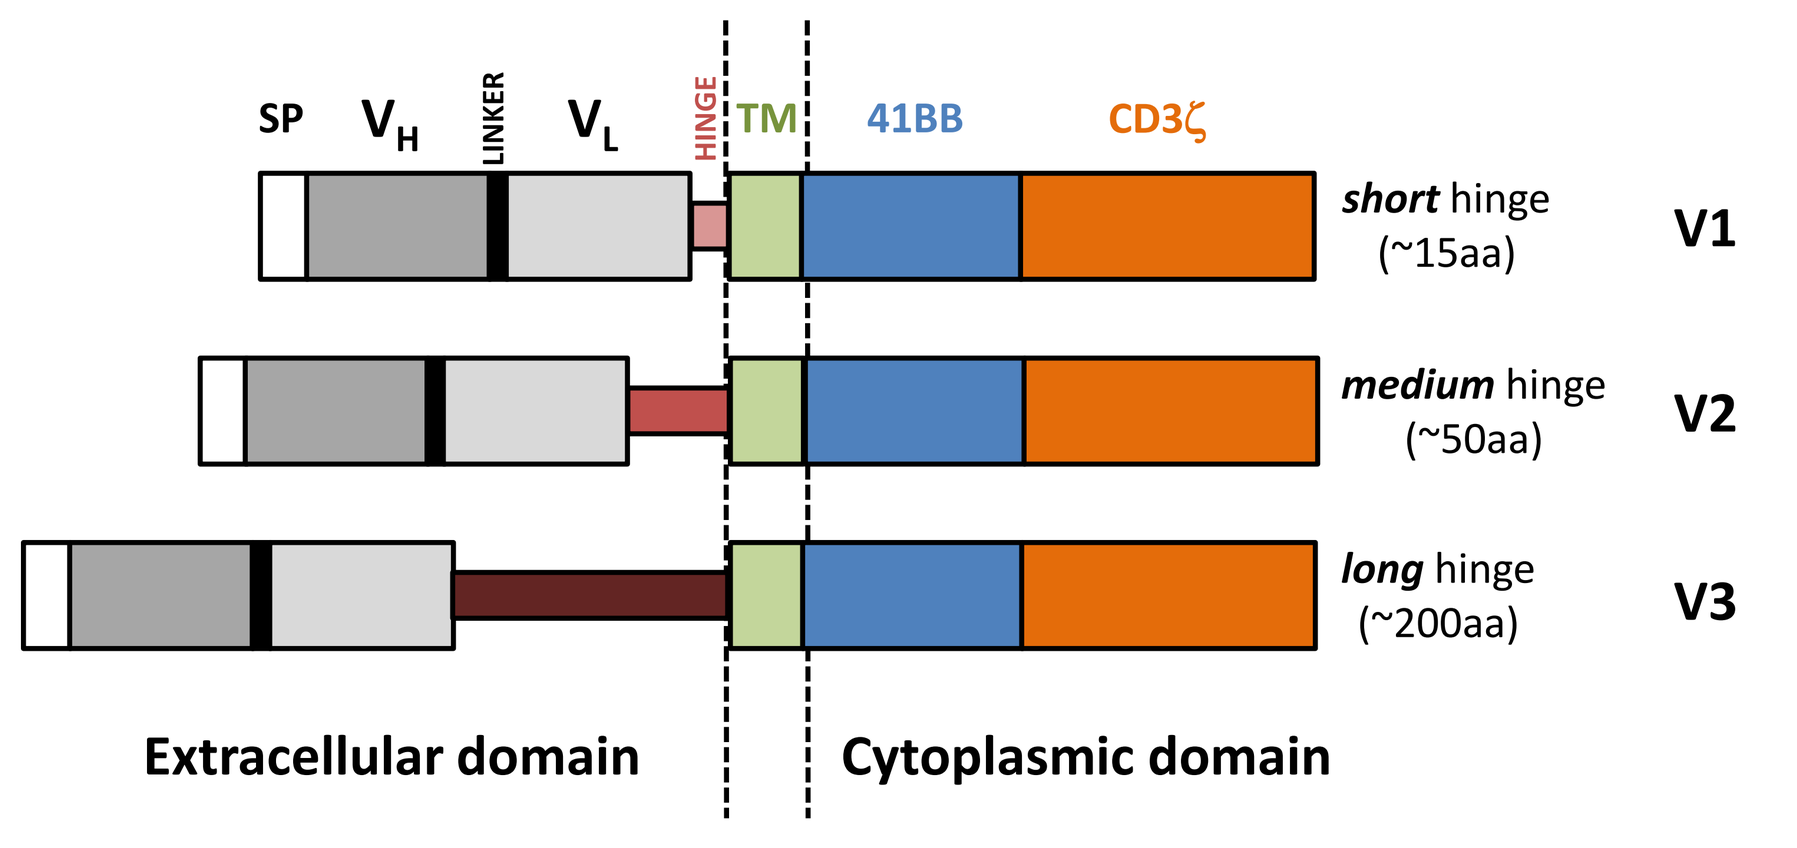

SP
VH
LINKER
VL
HINGE
TM
41BB
CD3ζ
short hinge (~15aa)
V1
medium hinge (~50aa)
V2
long hinge (~200aa)
V3
Extracellular domain
Cytoplasmic domain

#5 CD123 CAR T-cell in vitro activity

T-cells stably expressing the three CAR constructs and a benchmark CAR (32716) were generated by lentiviral transduction and their activity was assessed 10 days after transduction.

DEGRANULATION ASSAY

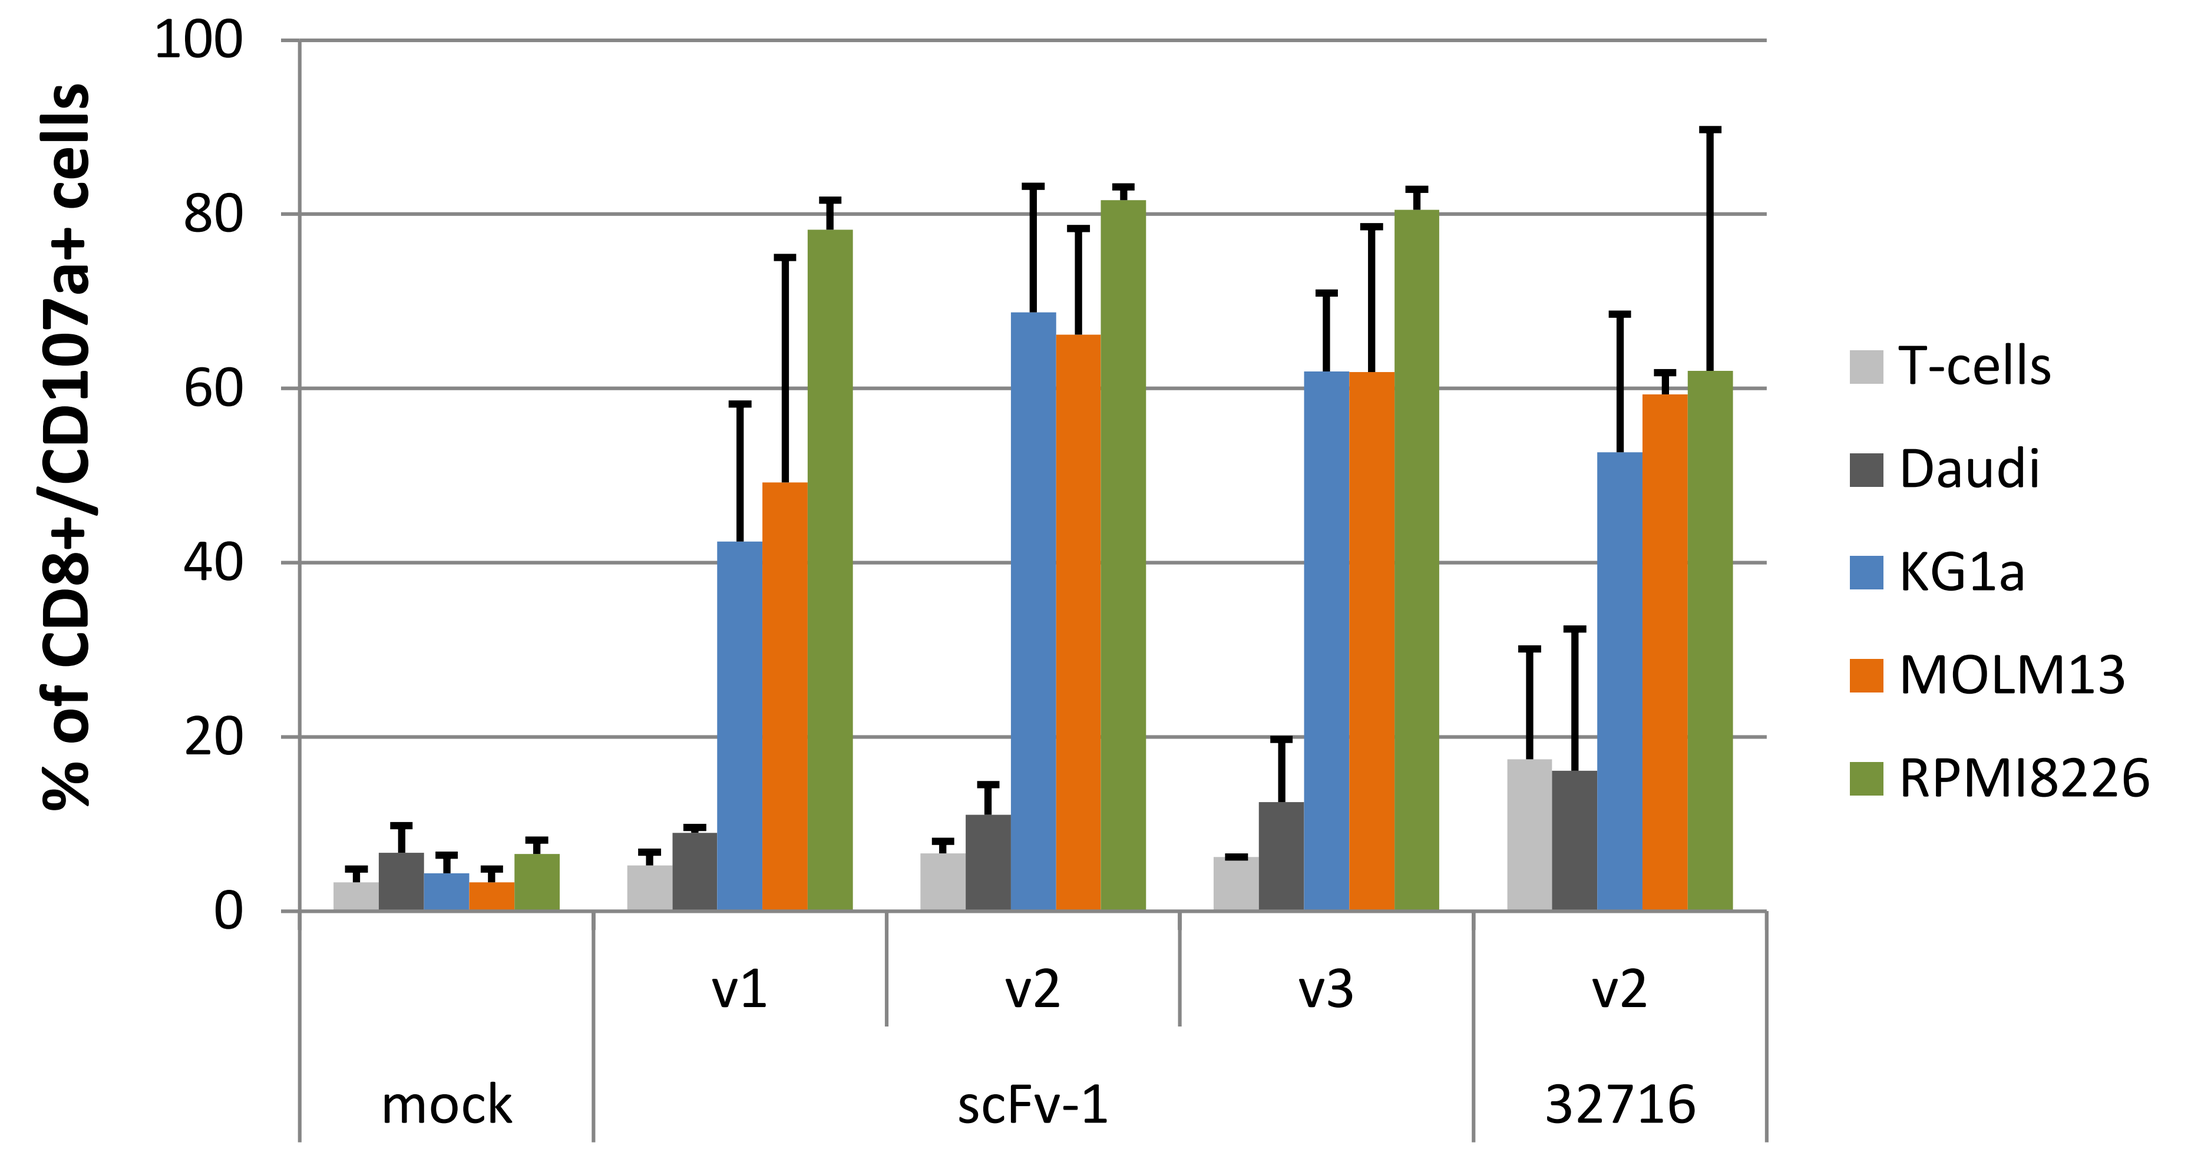

% of CD8+/CD107a+ cells
T-cells
Daudi
KG1a
MOLM13
RPMI8226
mock
v1
v2
v3
scFv-1
v2
32716

CAR+ T-cells were co-cultured with CD123+ target cells (1:1). Degranulation activity was measured by staining with CD8 and CD107a antibodies and analyzed by flow cytometry. Daudi cells were used as a negative control.

IFNgamma RELEASE ASSAY

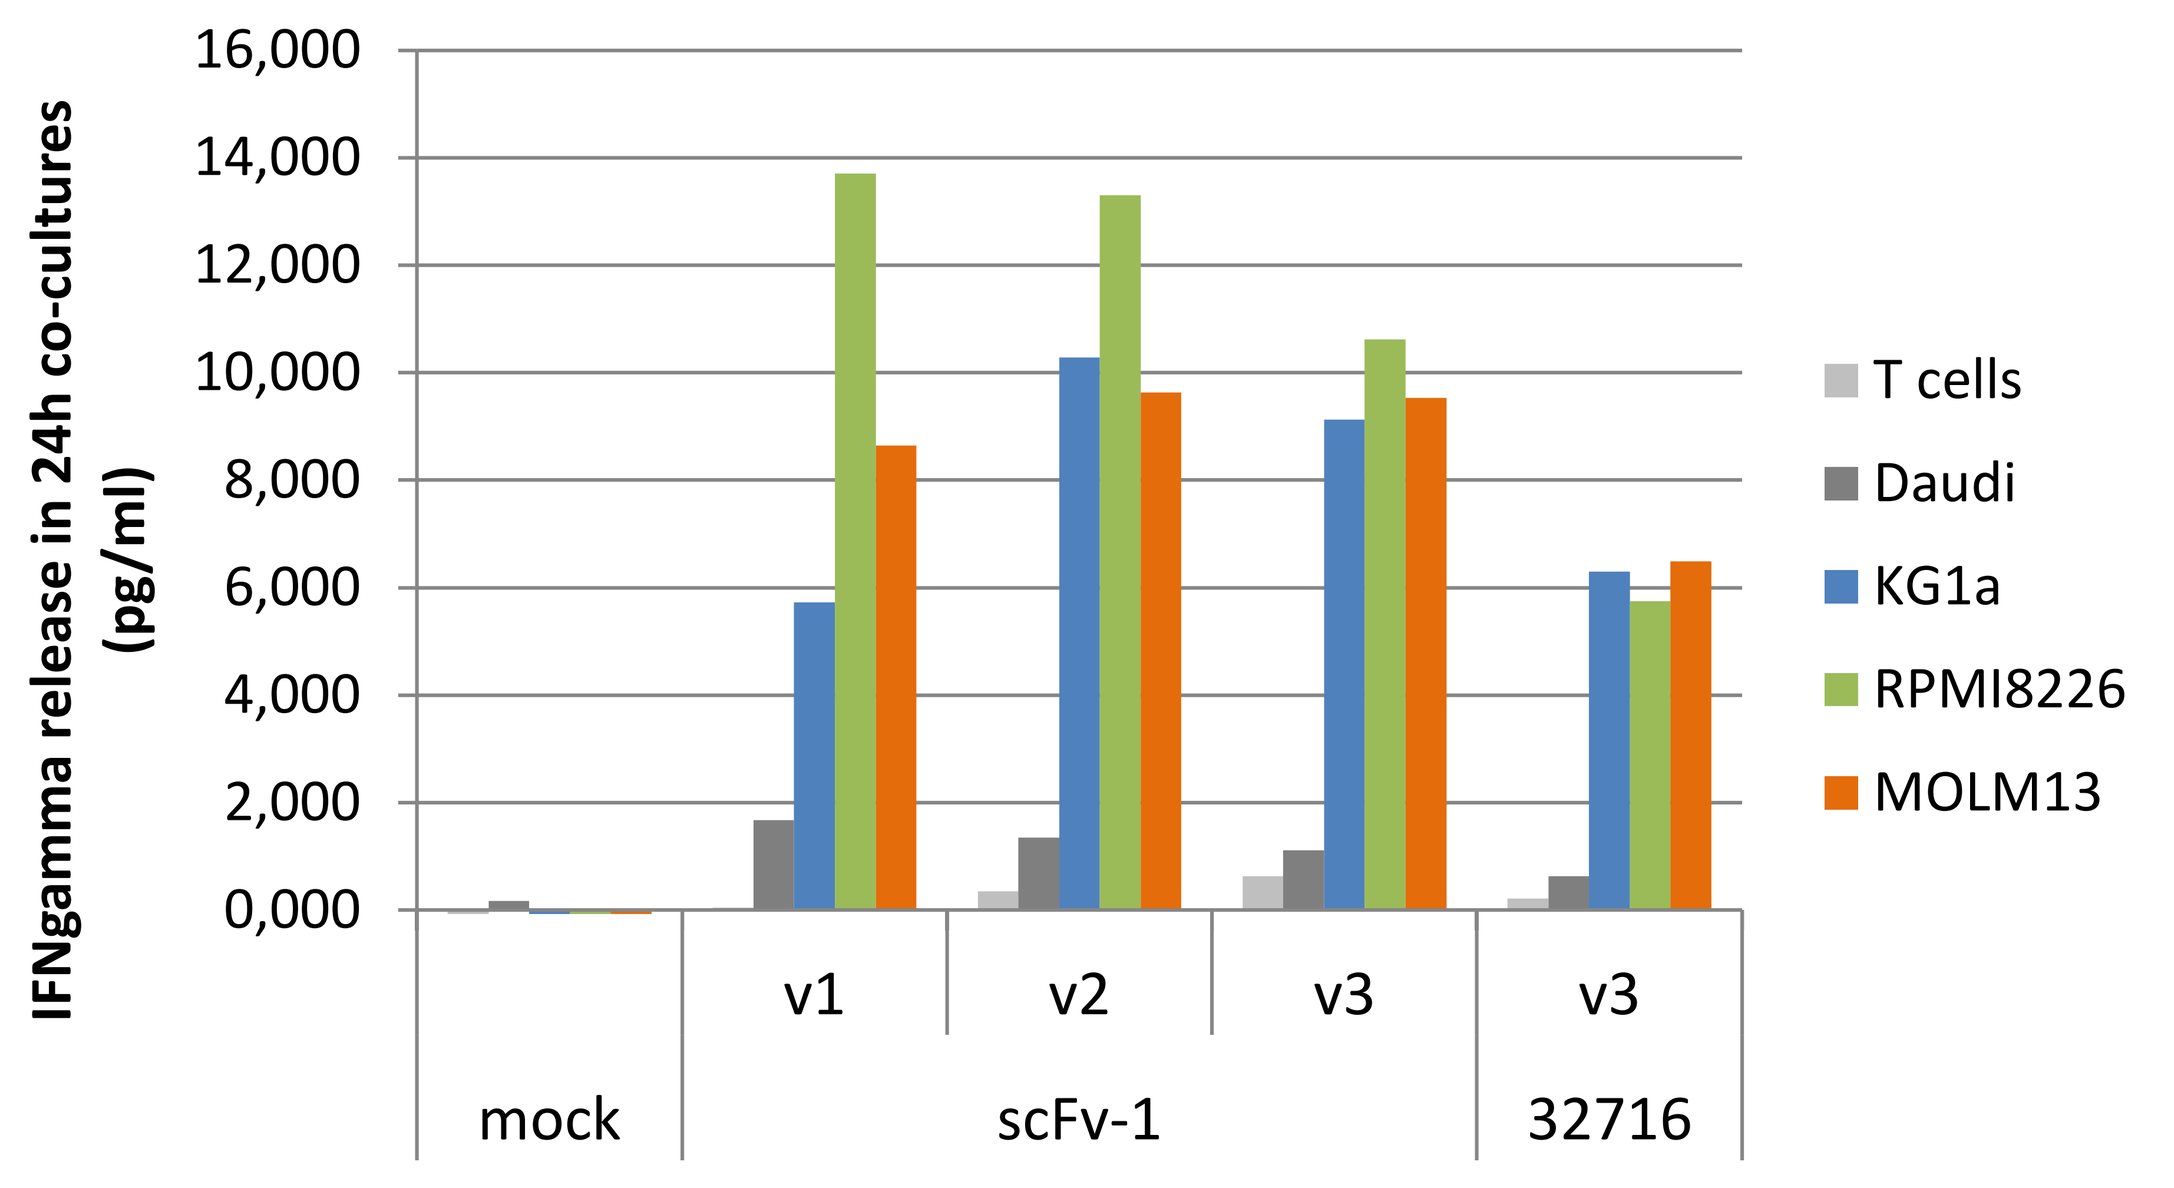

IFNgamma release in 24h co-cultures (pg/ml)
T cells
Daudi
KG1a
RPMI8226
MOLM13
mock
v1
v2
v3
scFv-1
v3
32716

CAR+ T-cells were co-cultured with CD123(+) target cells (1:1). The amount of IFNg released in the culture supernatants was determined by ELISA.

CYTOTOXICITY ASSAY

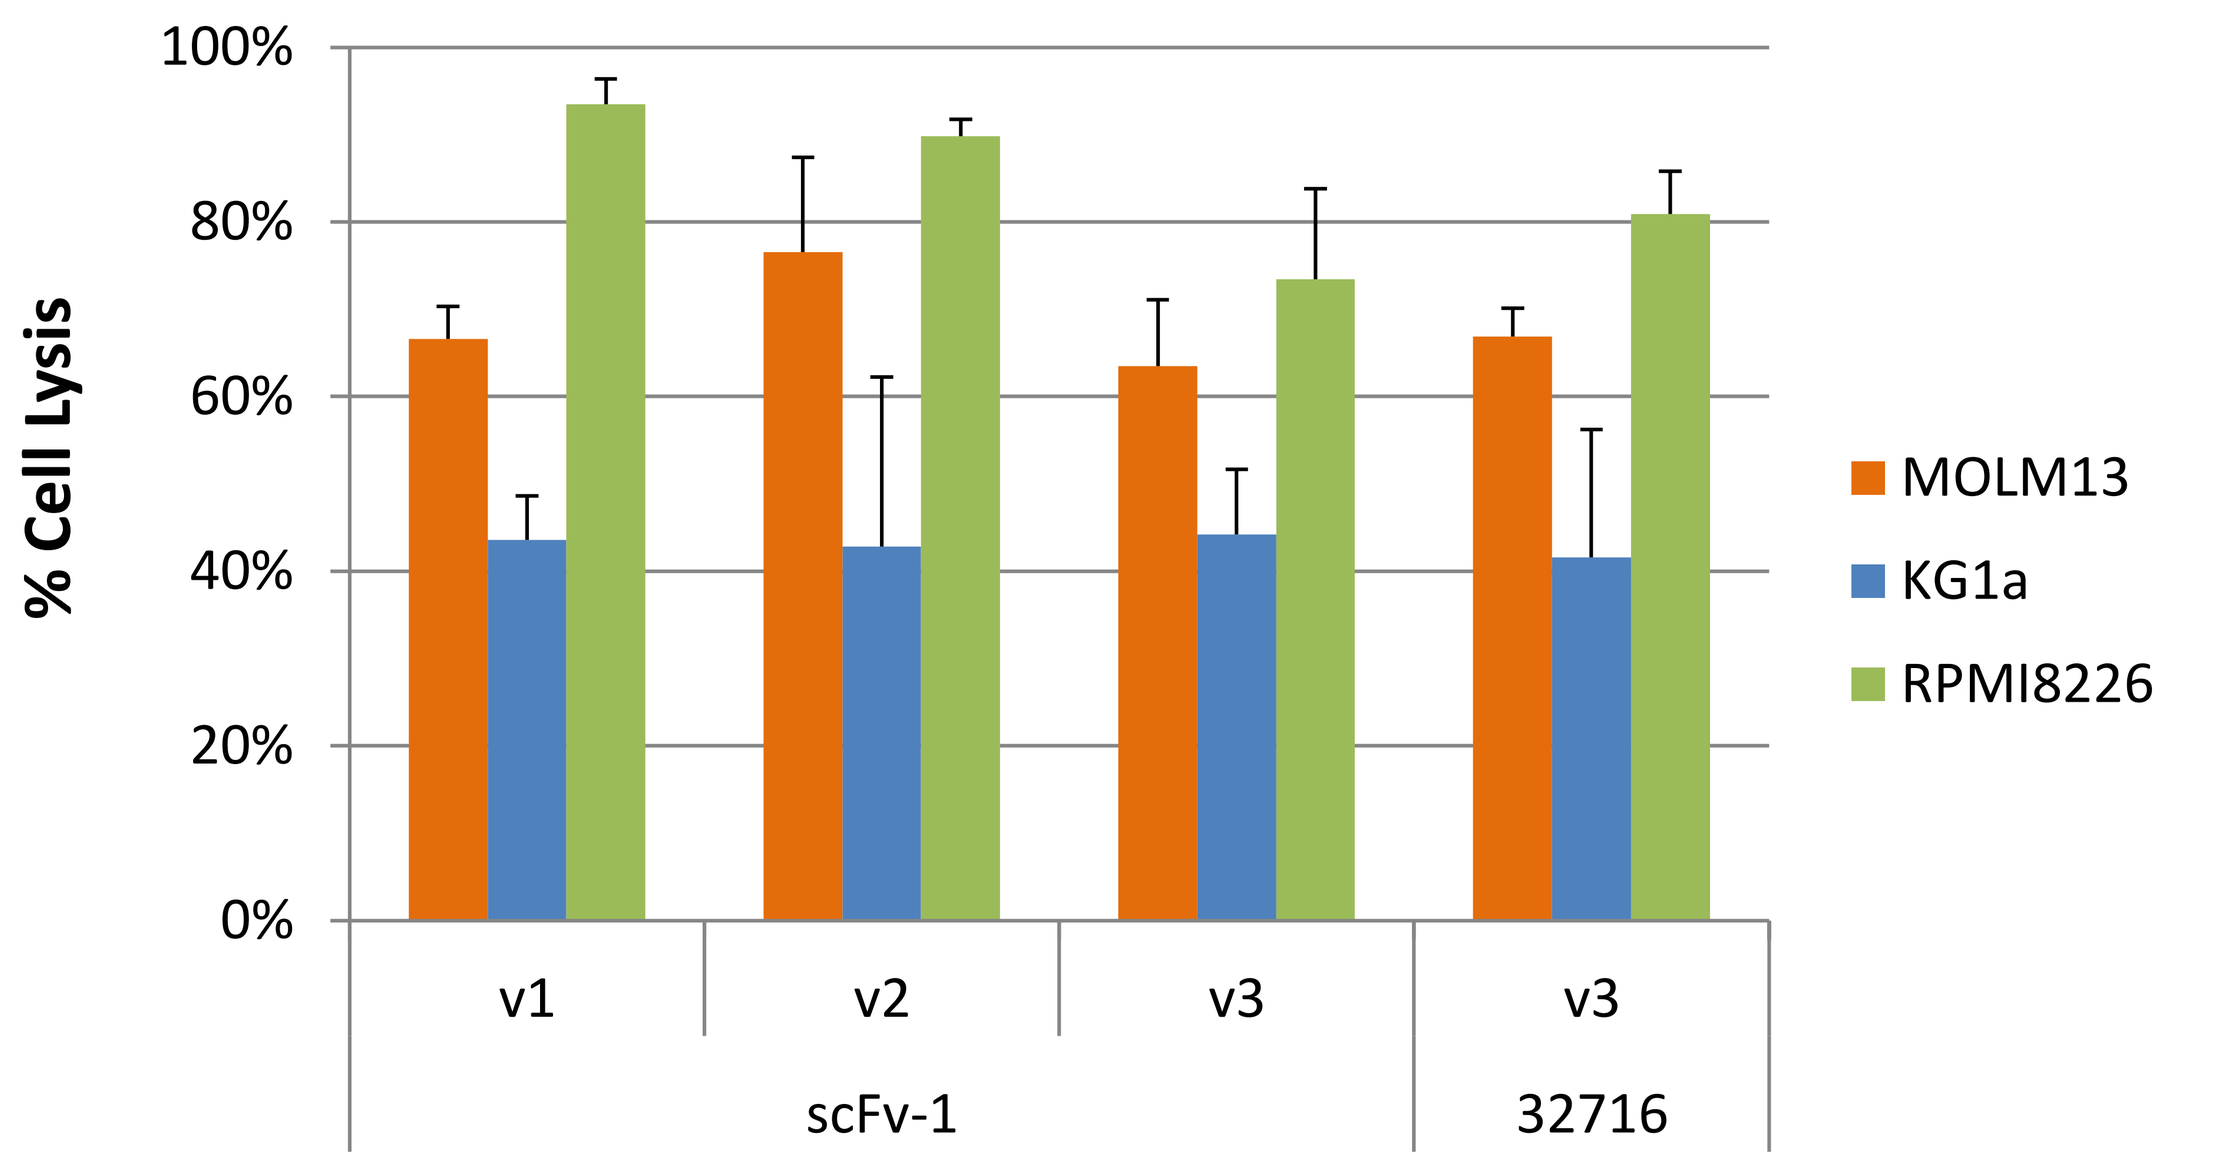

% Cell Lysis
MOLM13
KG1a
RPMI8226
v1
v2
v3
scFv-1
v3
32716

CAR+ T-cells were co-cultured with CD123(+) target cells and CD123(-) cells (10:1:1 ratio). The viability of each cell line -CD123(+) and CD123(-)- was determined after 4h of co-culture to determine the % of specific cell lysis.

#6 CD123 CAR T cell in vivo anti-tumor activity

All three CAR constructs were evaluated for vivo anti-tumor activity: 5×10^6 CAR-positive T-cells were intravenously injected in NSG mice 7 days after tumor cell injection (2.5×10^5 Molm13-ffluc cells).

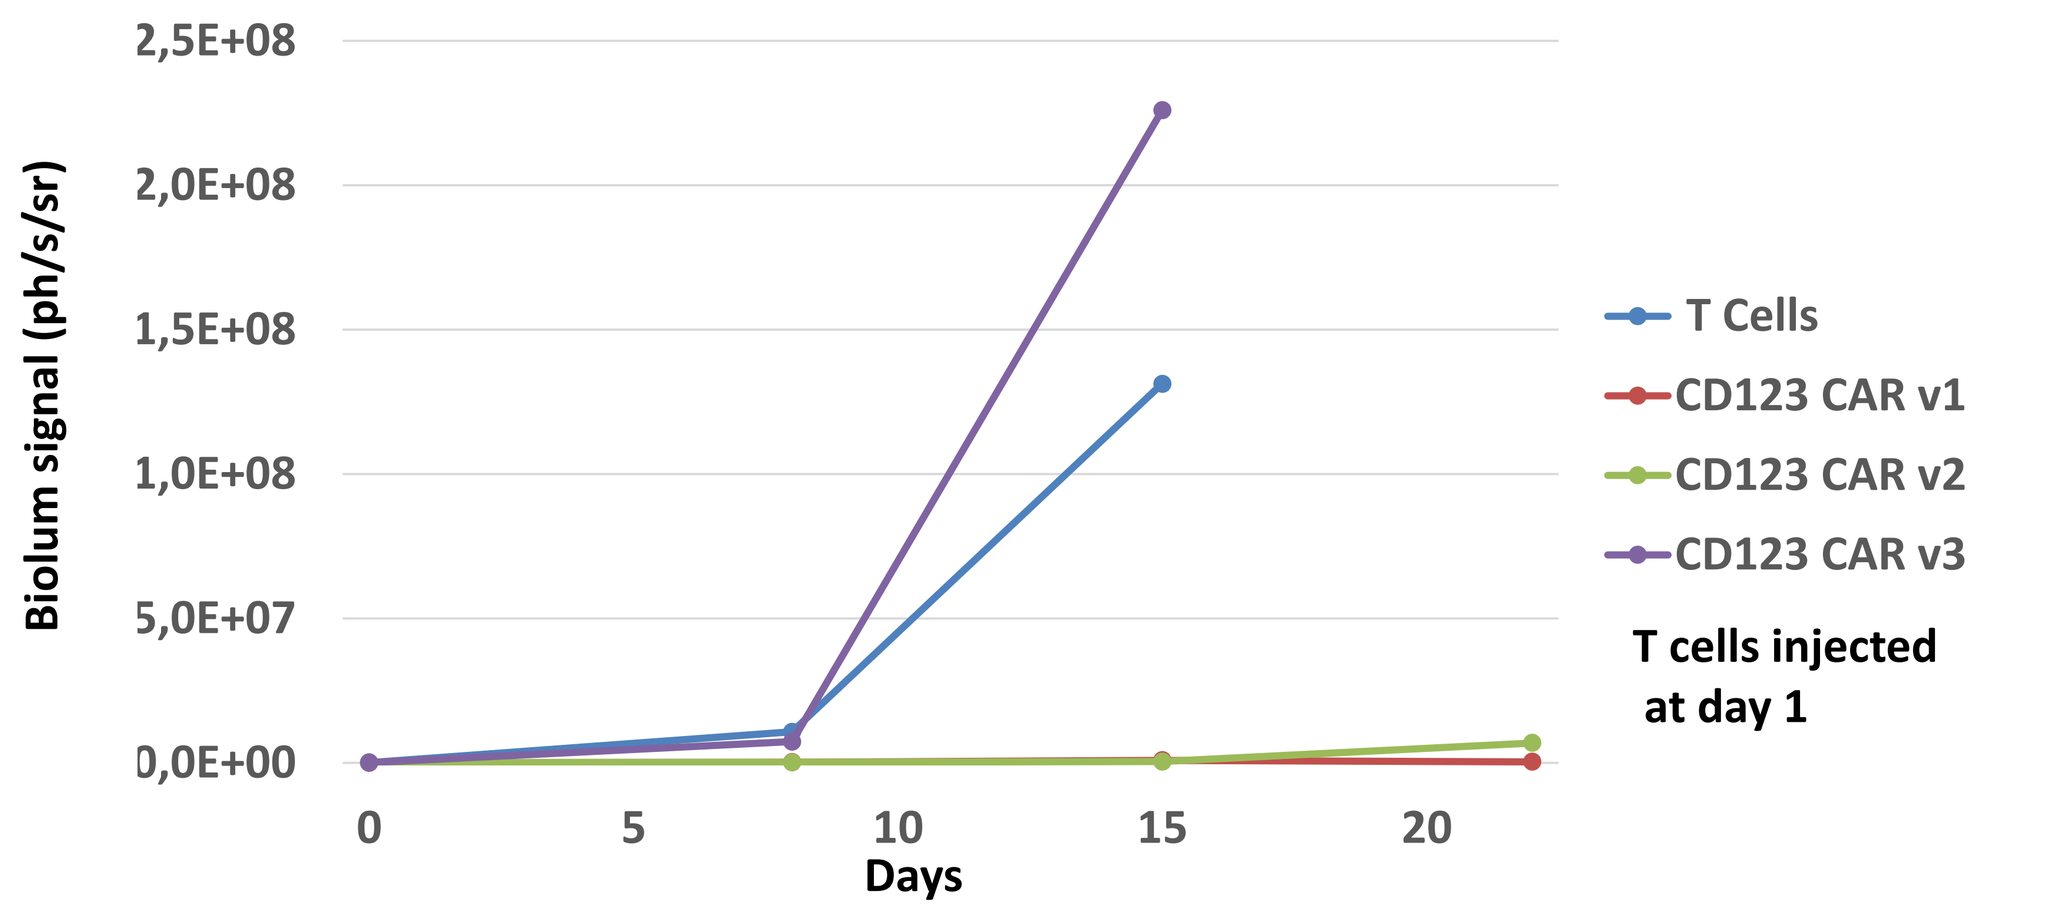

Biolum signal (ph/s/sr)
Days
T Cells
CD123 CAR v1
CD123 CAR v2
CD123 CAR v3
T cells injected at day 1

The CD123 CAR v1 and v2 display a strong anti-tumor effect while the CD123 CAR v3, despite significant in vitro activity, did not display any anti-tumor activity in vivo.

The anti-tumor activity of the v2 CAR was further characterized in a long term anti-tumor study: several doses of CAR-positive T-cells (1×10^6, 5×10^6 and 10×10^6 cells) were intravenously injected in NSG mice 7 days after tumor cell injection (2.5×10^5 Molm13-ffluc cells) and mice were followed for up to 60 days. The T cells were injected at Day 1 in the figures below.

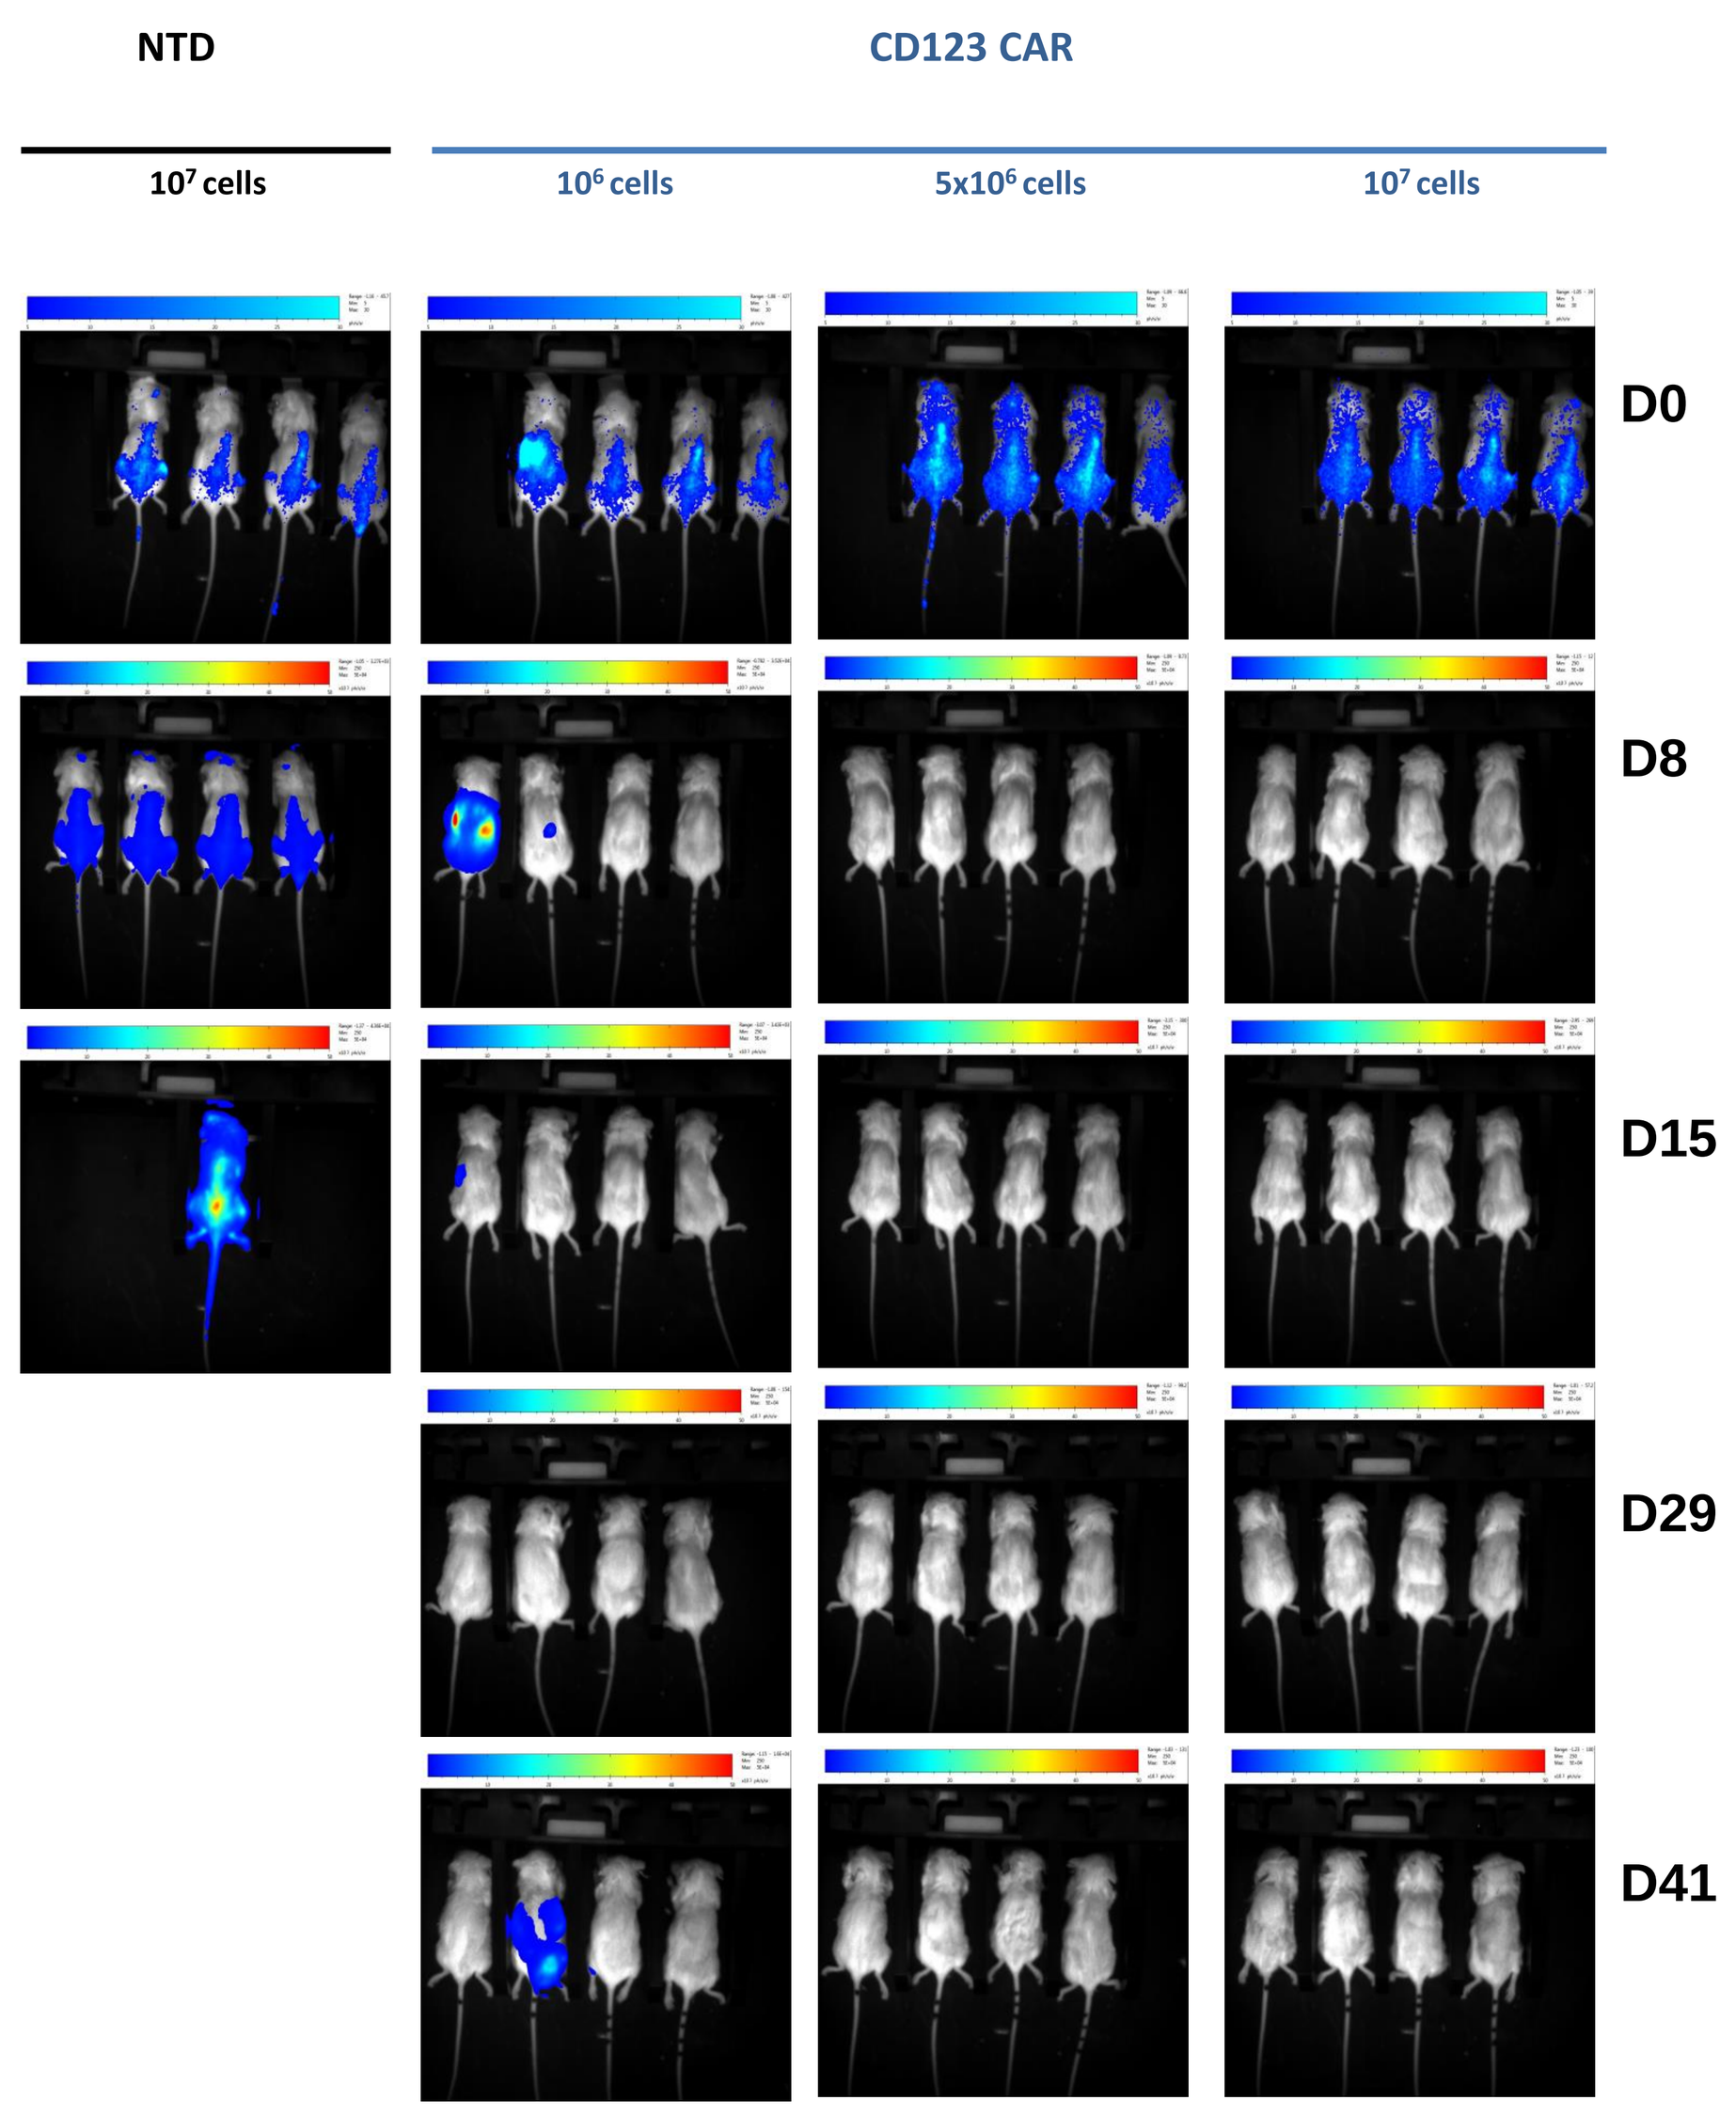

NTD
CD123 CAR
10⁷ cells
10⁶ cells
5x10⁶ cells
10⁷ cells
D0
D8
D15
D29
D41

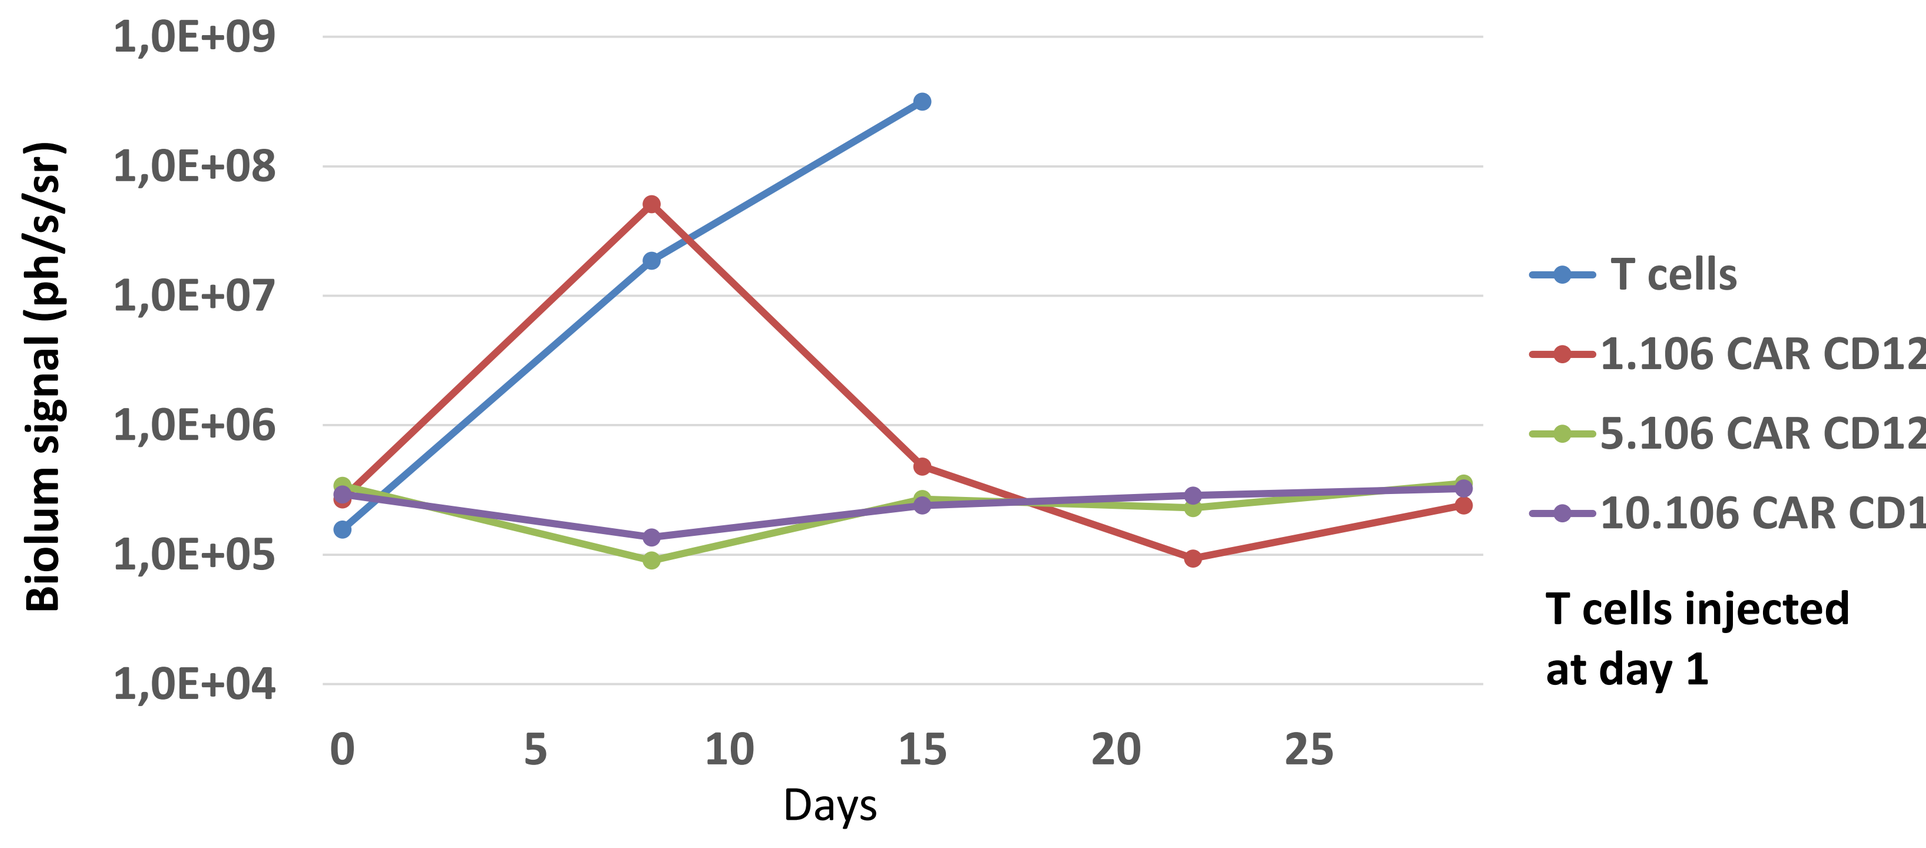

Biolum signal (ph/s/sr)
Days
T cells
1.106 CAR CD123
5.106 CAR CD123
10.106 CAR CD123
T cells injected at day 1

Except for one mouse (treated at 1x10^6), all the mice treated with CAR T-cells were alive at Day 60. In addition, flow cytometry analysis at Day 60 confirmed the complete elimination of the tumor cells in the blood, the spleen and the bone marrow.

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis and its business. Cellectis is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. Cellectis proprietary information

UCART19, an Allogeneic “Off-the-Shelf” Adoptive T-cell Immunotherapy against CD19+ B-cell Leukemias

Julianne Smith[1], Laurent Poirot[1], Brian Philip[2], Cécile Schiffer Mannioui[1], Sophie Derniame[1], Hong Zhan[2], Sian Stafford[2], Stephan Reynier[1], Sylvain Arnould[1], Agnès Gouble[1], Waseem Qasim[2], Karl Peggs[2], Martin Pule[2] and David Sourdive[1]

[1]Cellectis, Paris, France
[2]University College London, London, UK

#1 Abstract

Background: Autologous T-cells engineered to express chimeric antigen receptors (CARs) that target specific tumor antigens are of high potential in treating different kinds of cancer. However, they must be generated on a “per patient” basis, thereby limiting the population of patients that could benefit from this approach.
Methods: We have developed a standardized platform for manufacturing T-cells from third-party healthy donors to generate allogeneic "off-the-shelf" engineered CD19-CAR+ T-cell–based frozen products. Our platform involves the use of transcription activator-like effector nucleases (TALEN), which mediate the simultaneous inactivation of two genes through genome editing. The knockout of the TCR alpha gene eliminates TCR expression and is intended to abrogate the donor T-cell’s potential for graft-versus-host disease (GvHD), while knocking out the CD52 gene makes donor T-cells resistant to the lymphodepleting agent alemtuzumab. In addition, our T-cells are engineered to co-express the RQR8 gene as a safety feature, with the aim of rendering them sensitive to the monoclonal antibody rituximab.
Results: We have obtained proof-of-concept by manufacturing TCR/CD52-deficient RQR8+ and CD19-CAR+ T-cells (UCART19) using a good manufacturing practice–compatible process and have demonstrated that UCART19 cells were functional using in vitro assays. Furthermore, we have demonstrated that the ability of UCART19 cells to engraft into an orthotopic human CD19+ lymphoma xenograft immunodeficient mouse model. UCART19 cells exhibited antitumor activity equivalent to that of standard CD19 CAR T-cells. We also demonstrated that UCART19 cells did not mediate alloreactivity in a xeno-GvHD mouse model. Finally, the effectiveness of the rituximab-induced depletion mechanism of RQR8+cells was shown in an immunocompetent mouse model.
Conclusions: This valuable dataset supports the development of allogeneic CAR T-cells, and UCART19 will be investigated in an exploratory, first-in-human, clinical trial where refractory/relapsed CD19+ B-cell leukemia patients are to be enrolled.

#2 UCART19: an allogeneic CAR T cell for ALL and CLL

Cell Development Process:

1. Start from healthy, unmodified donor T-Cells
2. Insertion of single-chain CAR construct and suicide gene
3. Gene-knockout of TCR alpha to avoid GvHD and CD52 for Alemtuzumab resistance

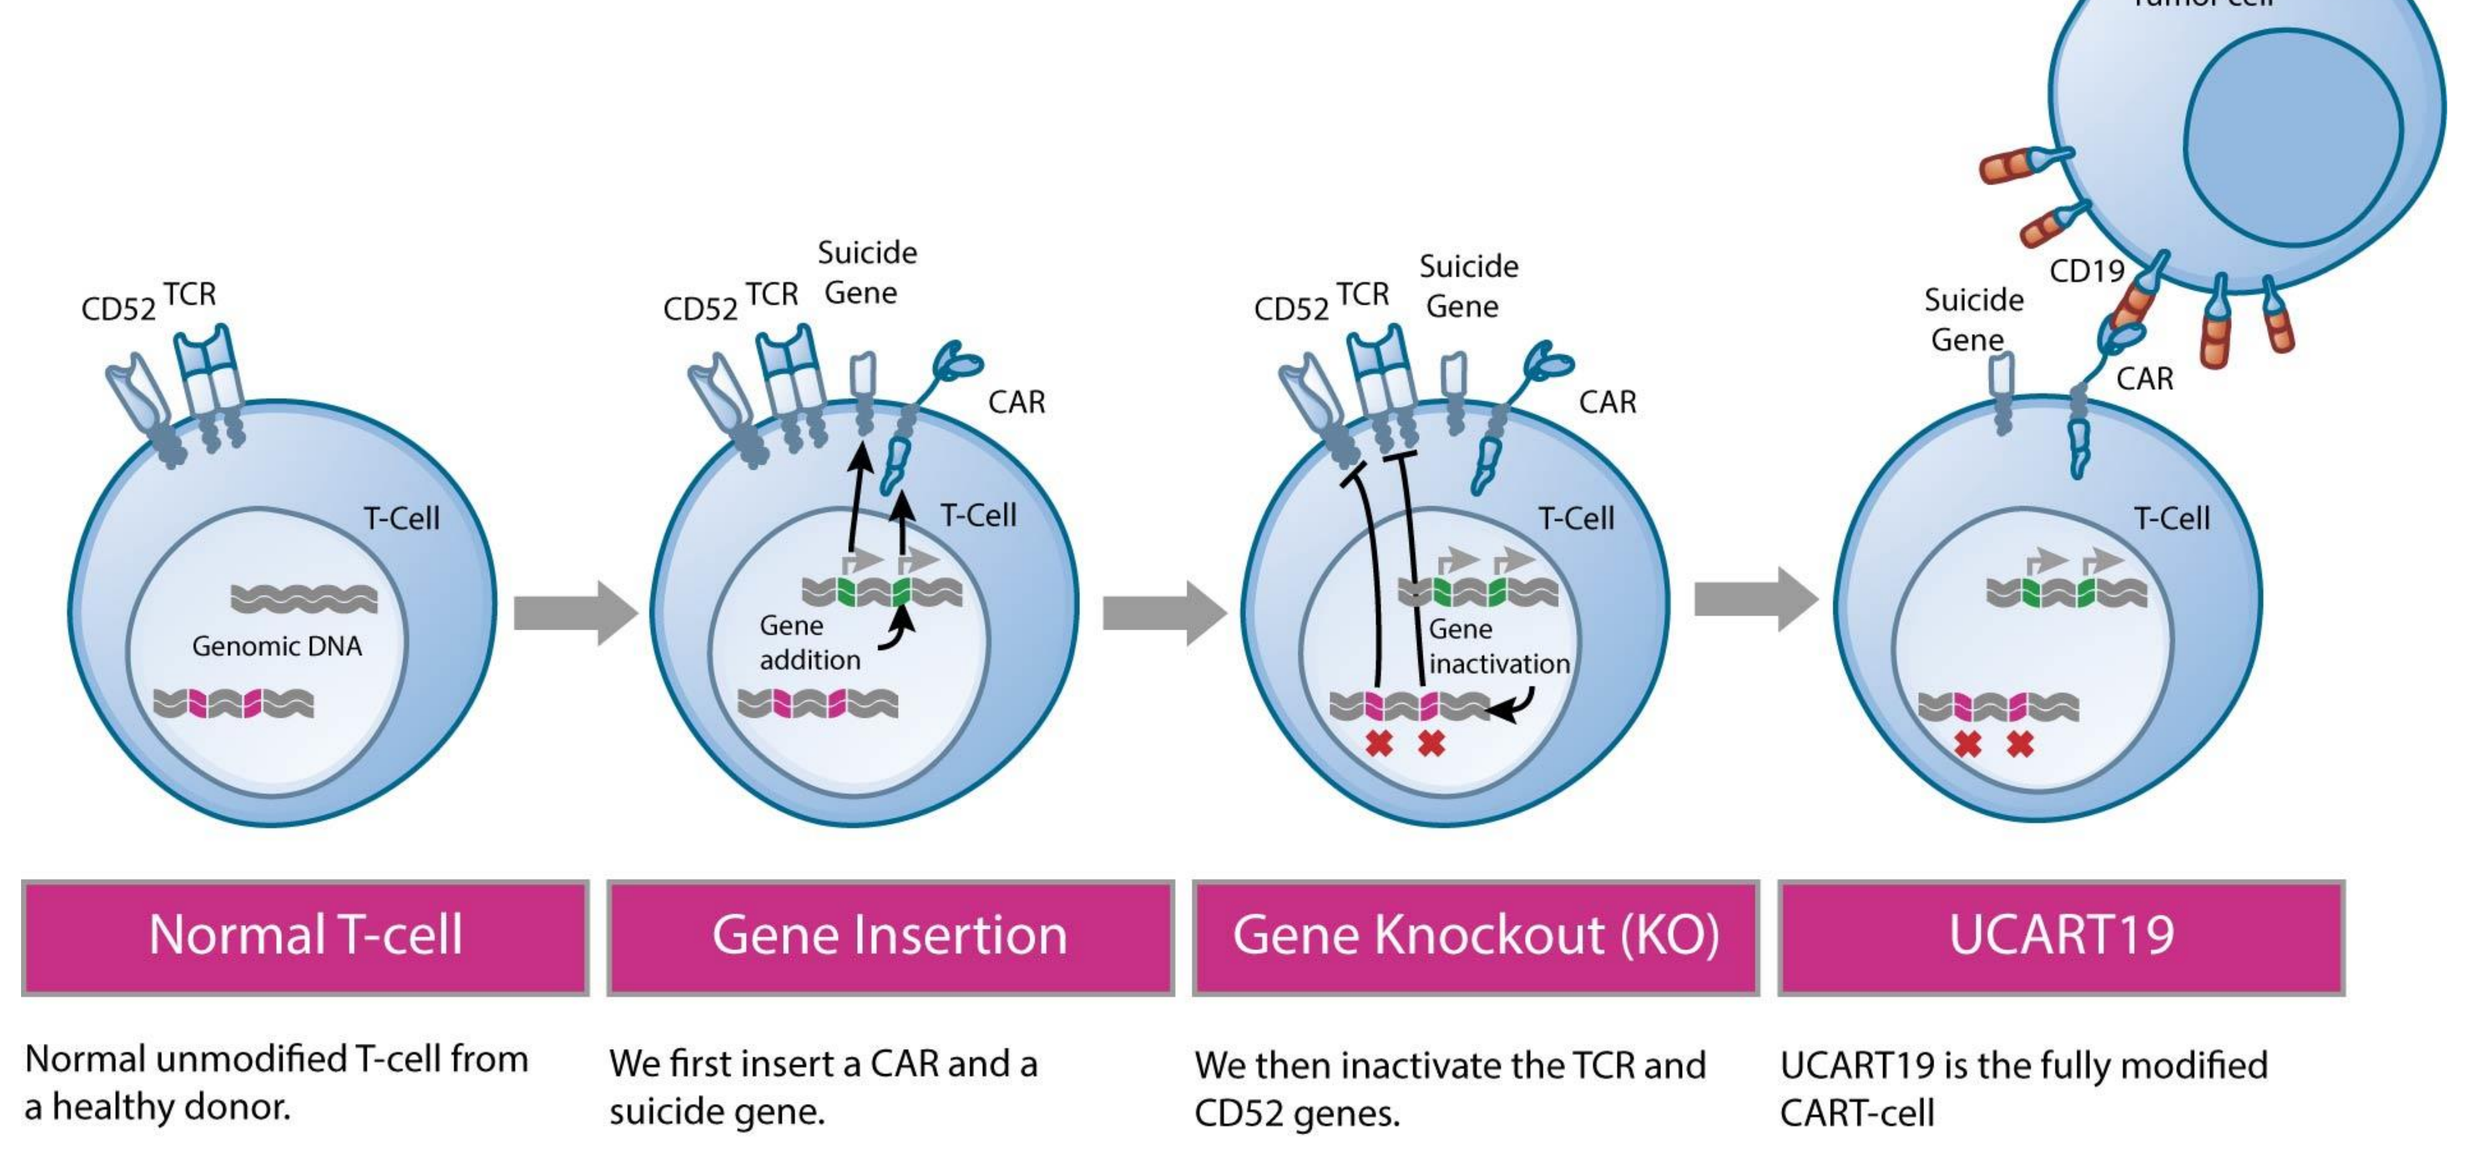
Tumor cell
CD52 TCR
Suicide Gene
CAR
CD19
T-Cell
Genomic DNA
Gene addition
Gene inactivation
Normal T-cell
Gene Insertion
Gene Knockout (KO)
UCART19
Normal unmodified T-cell from a healthy donor.
We first insert a CAR and a suicide gene.
We then inactivate the TCR and CD52 genes.
UCART19 is the fully modified CART-cell

The key barriers to the adoptive transfer of 3rd party CAR T-cells can be overcome via application of TALEN gene editing technology - deletion of the TCR gene allows use of any donors’ T-cells without the potential for GvHD, while deletion of CD52 allows for resistance to the lymphodepleting agent alemtuzumab.

#3 Optimized UCART19 T cell Manufacturing Process

Cellectis has developed and optimized a 17 day T-cell manufacturing process permitting production of TCR/CD52-deficient CAR T-cells with highly reproducible yields and cell phenotypes.

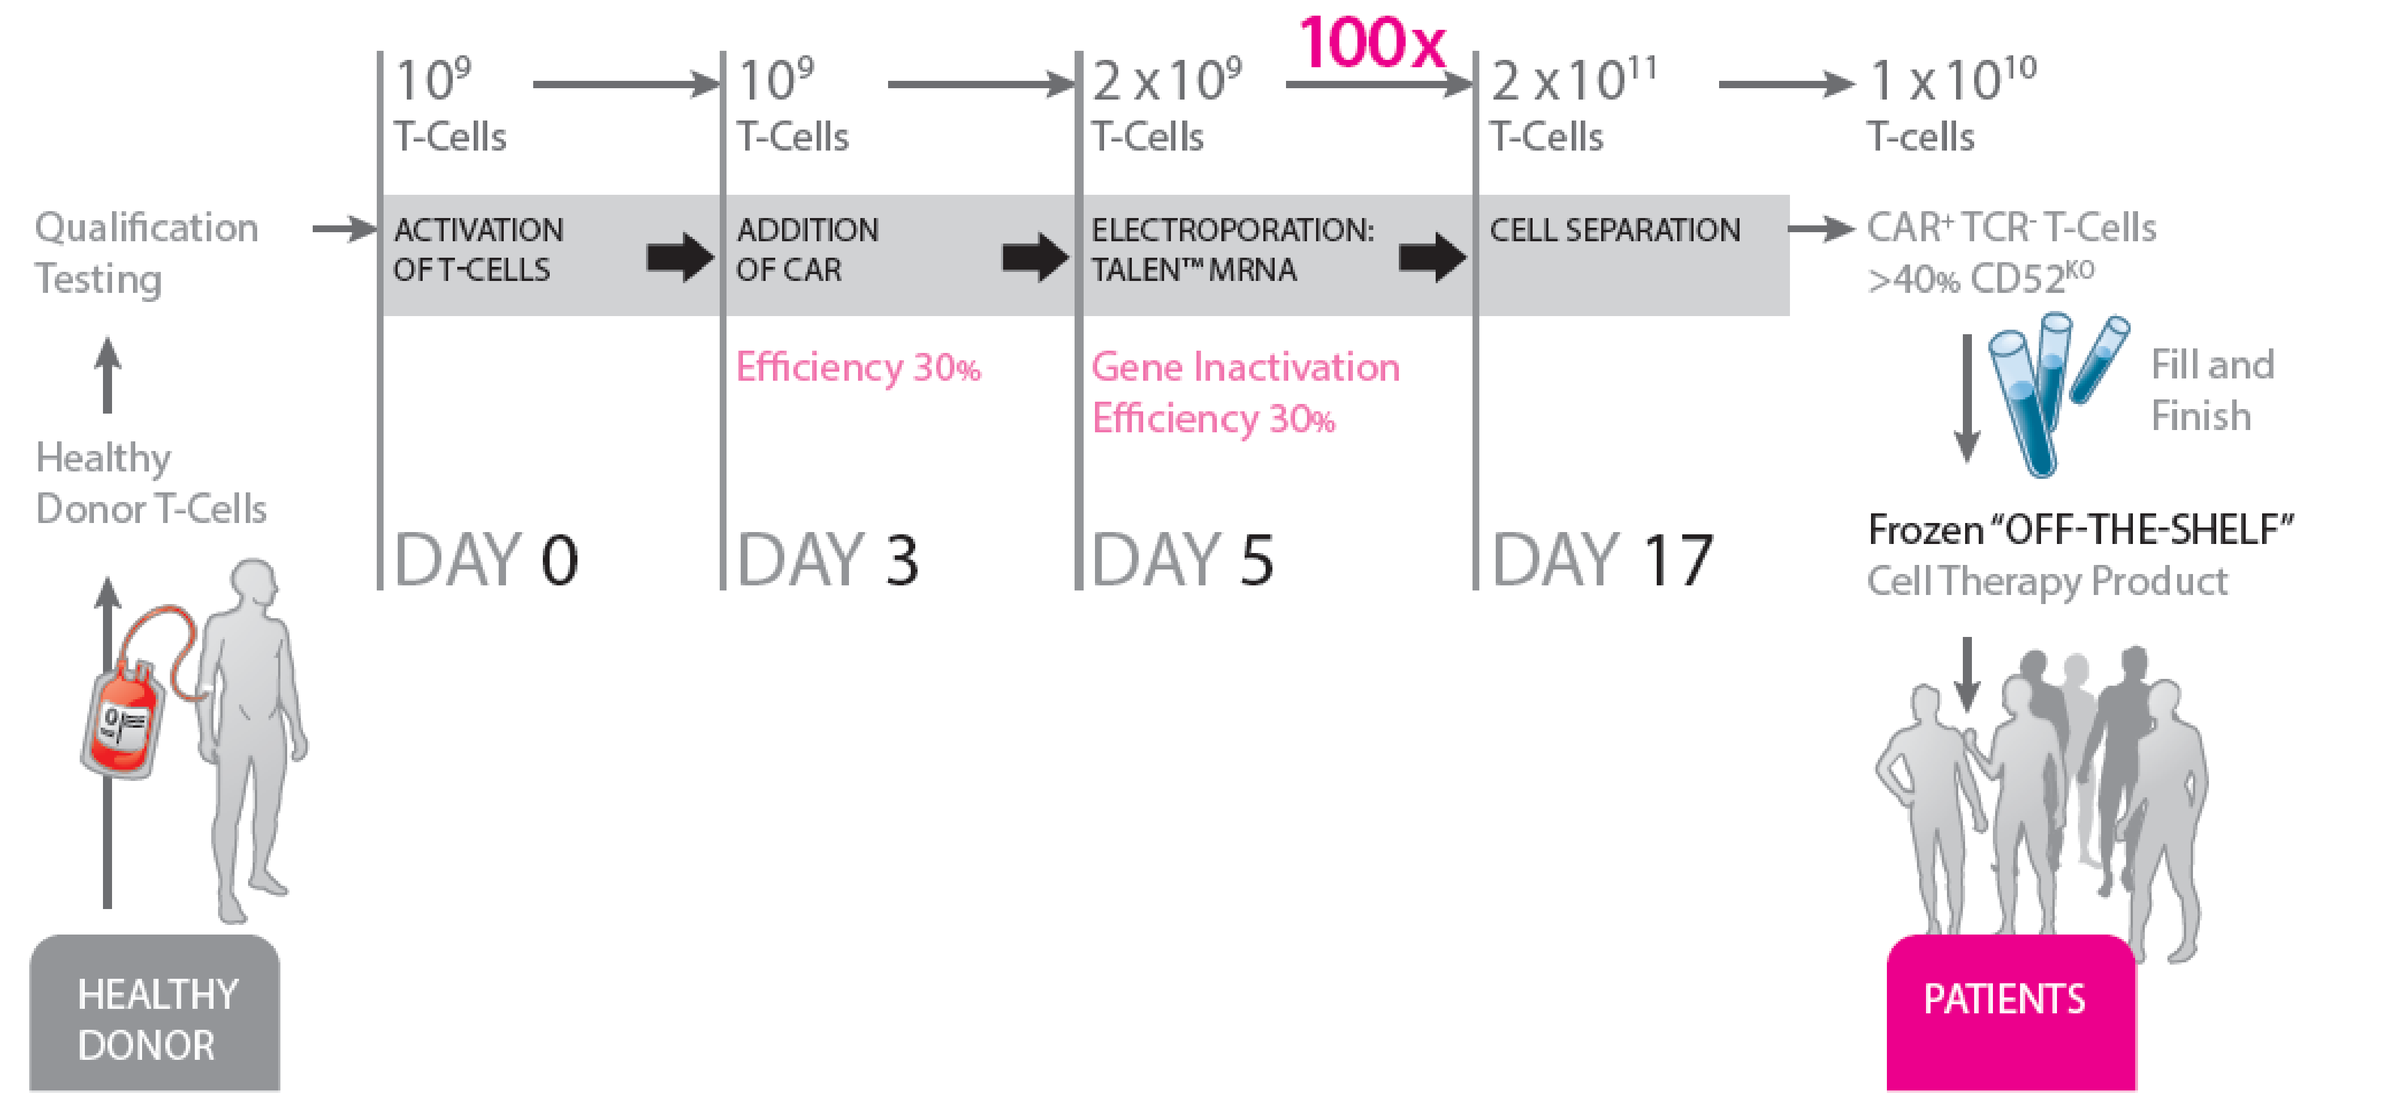
Healthy Donor T-Cells
Qualification Testing
HEALTHY DONOR
ACTIVATION OF T-CELLS
ADDITION OF CAR
ELECTROPORATION: TALEN™ MRNA
CELL SEPARATION
Efficiency 30%
Gene Inactivation Efficiency 30%
100x
DAY 0
DAY 3
DAY 5
DAY 17
CAR+ TCR- T-Cells >40% CD52KO
Fill and Finish
Frozen “OFF-THE-SHELF” Cell Therapy Product
PATIENTS

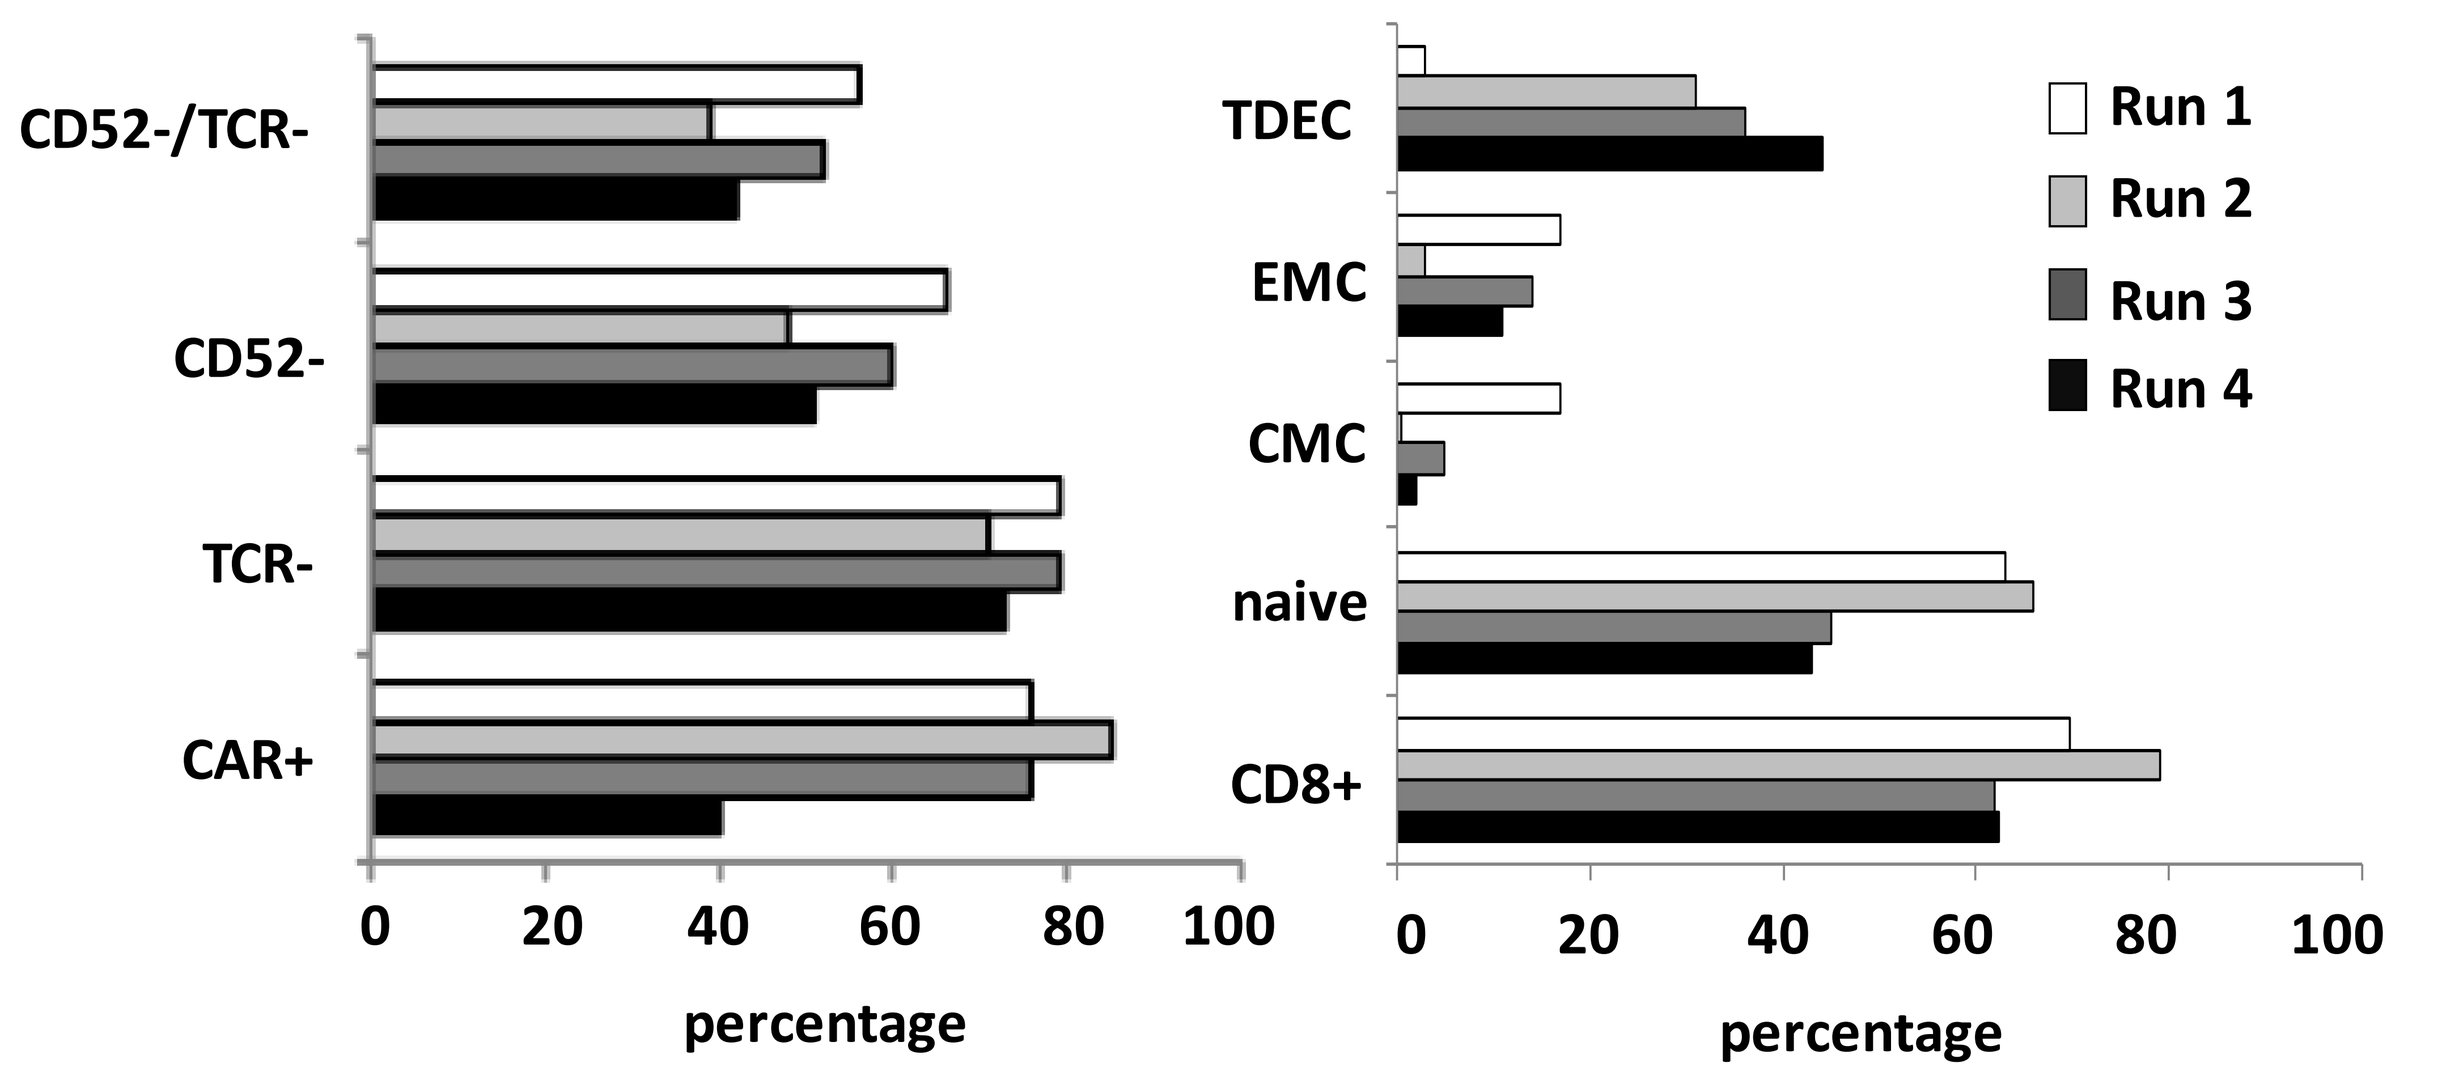
CD52-/TCR-
CD52-
TCR-
CAR+
TDEC
EMC
CMC
naive
CD8+
Run 1
Run 2
Run 3
Run 4
percentage

#4 UCART19 cells display potency equivalent to CAR T cells

Anti-tumor activity: 4×10^6 CAR-positive T-cells (UCART19 or regular CART19 cells) were intravenously injected in NSG mice either 1 day (5x10^5 Daudi-ffluc cells) or 7 days (2.5×10^5 Daudi-ffluc cells) after tumor cell injection. Mice were serially monitored for the first 3 weeks via luminescence imaging to follow tumor progression, and over an 8 week period for survival.

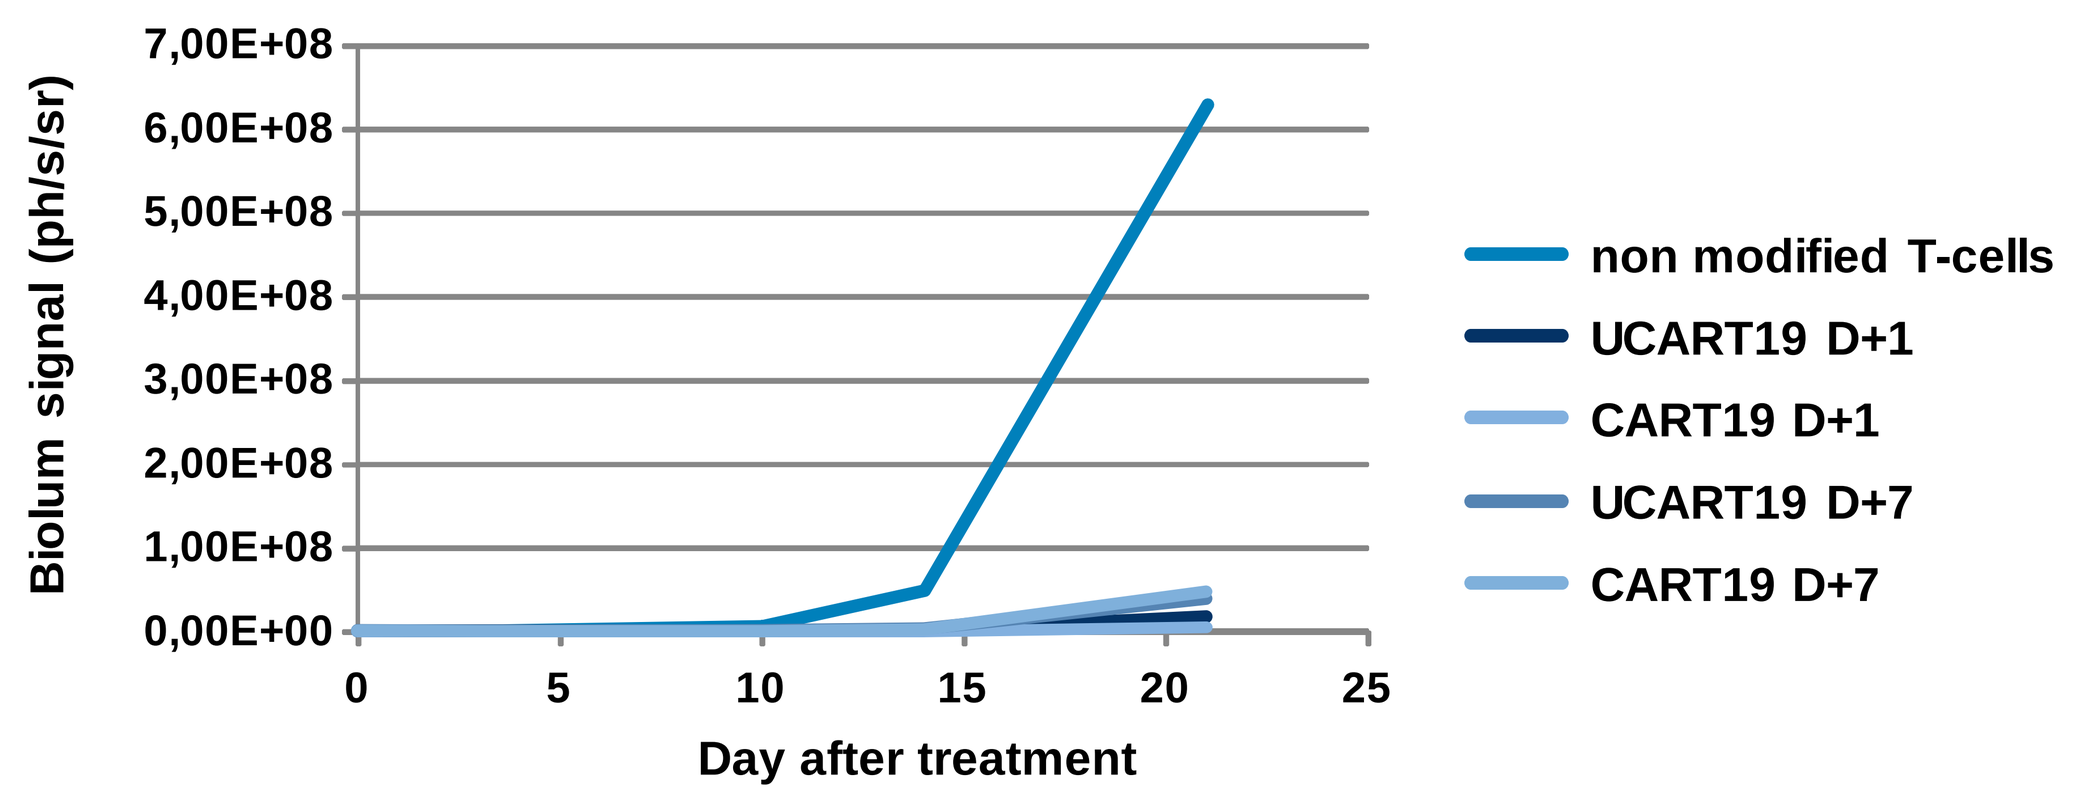
Biolum signal (ph/s/sr)
Day after treatment
non modified T-cells
UCART19 D+1
CART19 D+1
UCART19 D+7
CART19 D+7

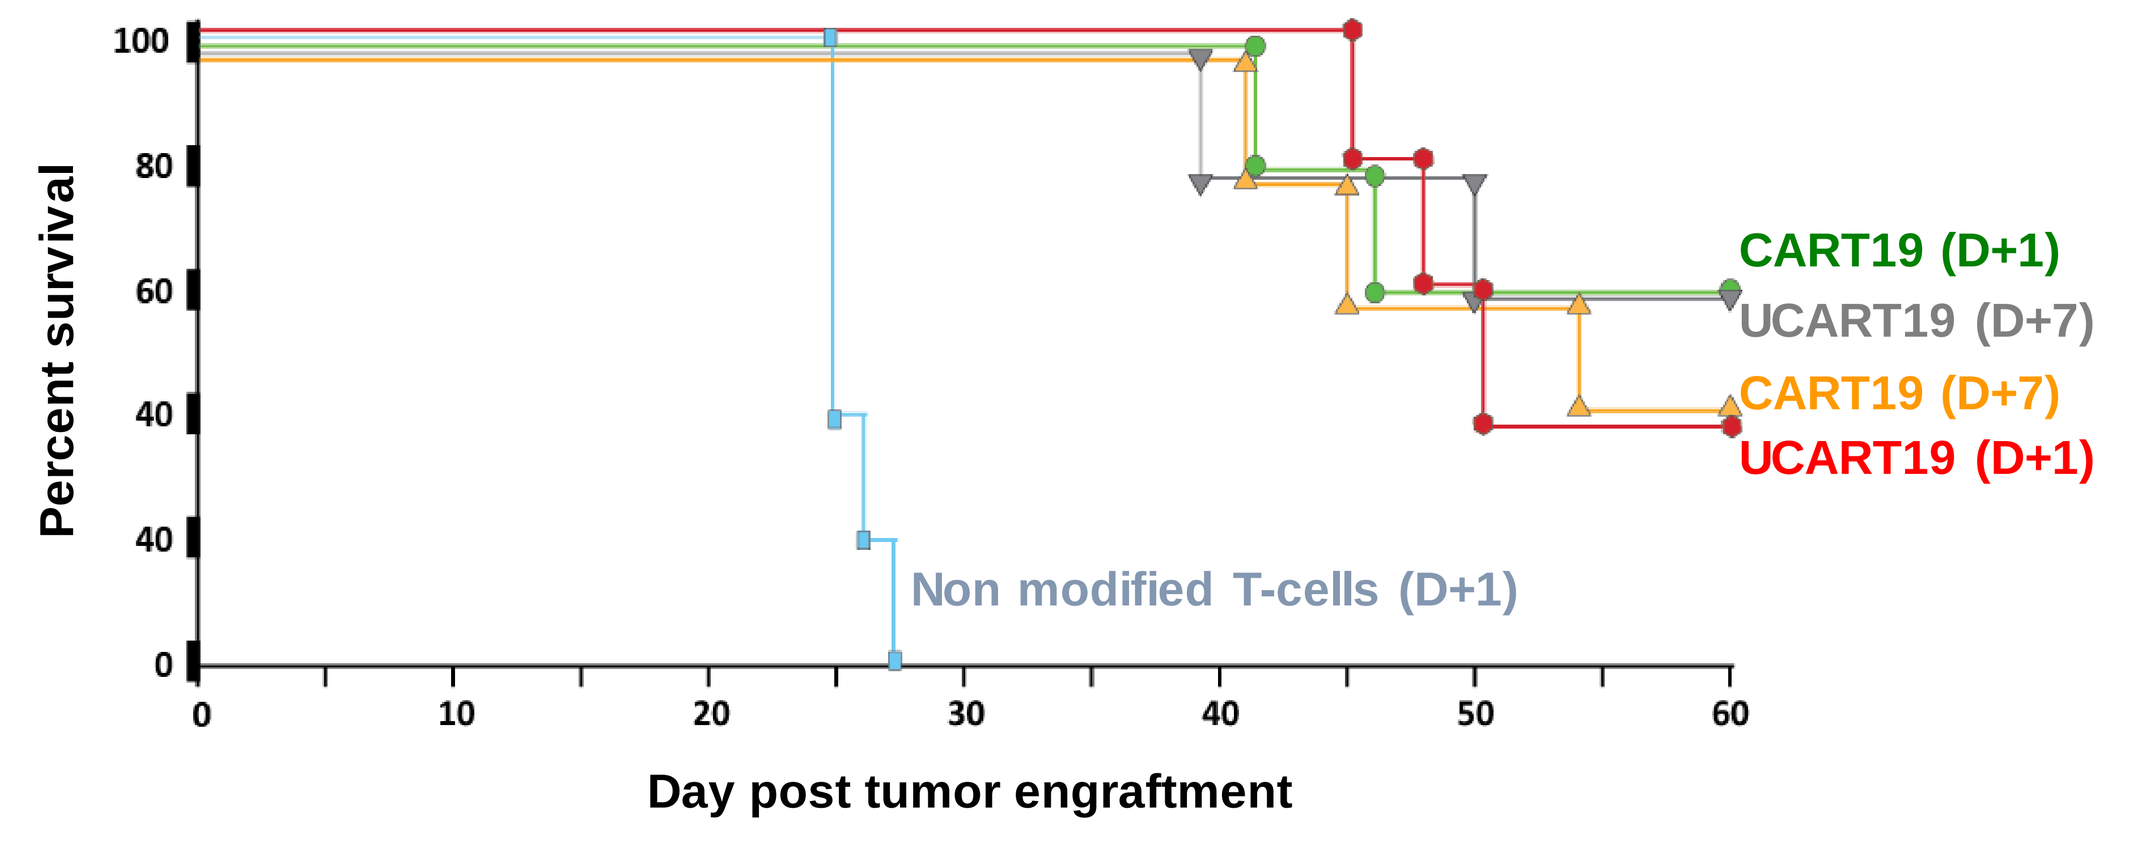
Percent survival
Day post tumor engraftment
CART19 (D+1)
UCART19 (D+7)
CART19 (D+7)
UCART19 (D+1)
Non modified T-cells (D+1)

UCART19 cells display a potency that is equivalent to standard CAR T cells indicating that the inactivation of TCR alpha and CD52 genes has no major impact on anti-tumor activity.

#5 TCR alpha inactivation prevents Graft vs. Host Disease

Irradiated NOG mice were injected with PBS (group 1), or 30x10^6 human T cells from the same donor. The cells were obtained from activated PBMCs after 3 day expansion (group 2), 15-day expansion (group 3), mock electroporation and 15-day expansion (group 4), or CD52/TRAC TALEN-electroporation, 15-day expansion and depletion of TCR+ cells (group 5).

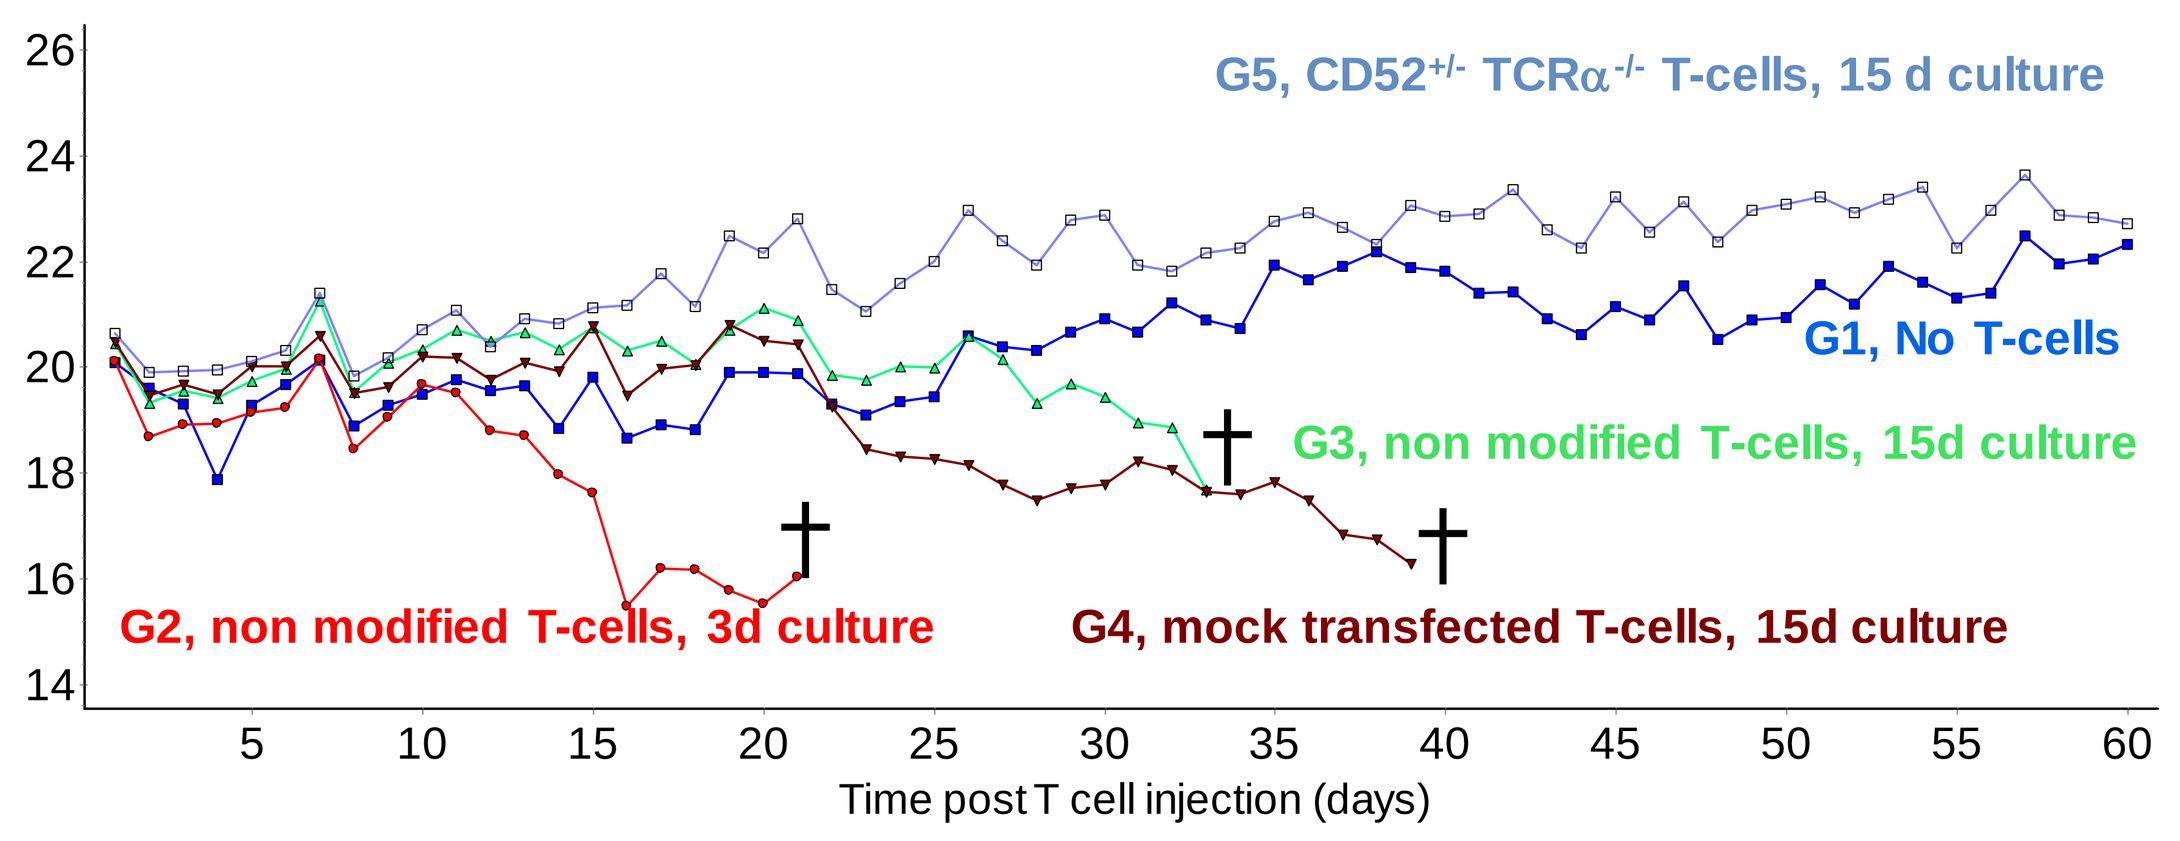
G5, CD52+/- TCRα-/- T-cells, 15 d culture
G1, No T-cells
G3, non modified T-cells, 15d culture
G2, non modified T-cells, 3d culture
G4, mock transfected T-cells, 15d culture
Time post T cell injection (days)

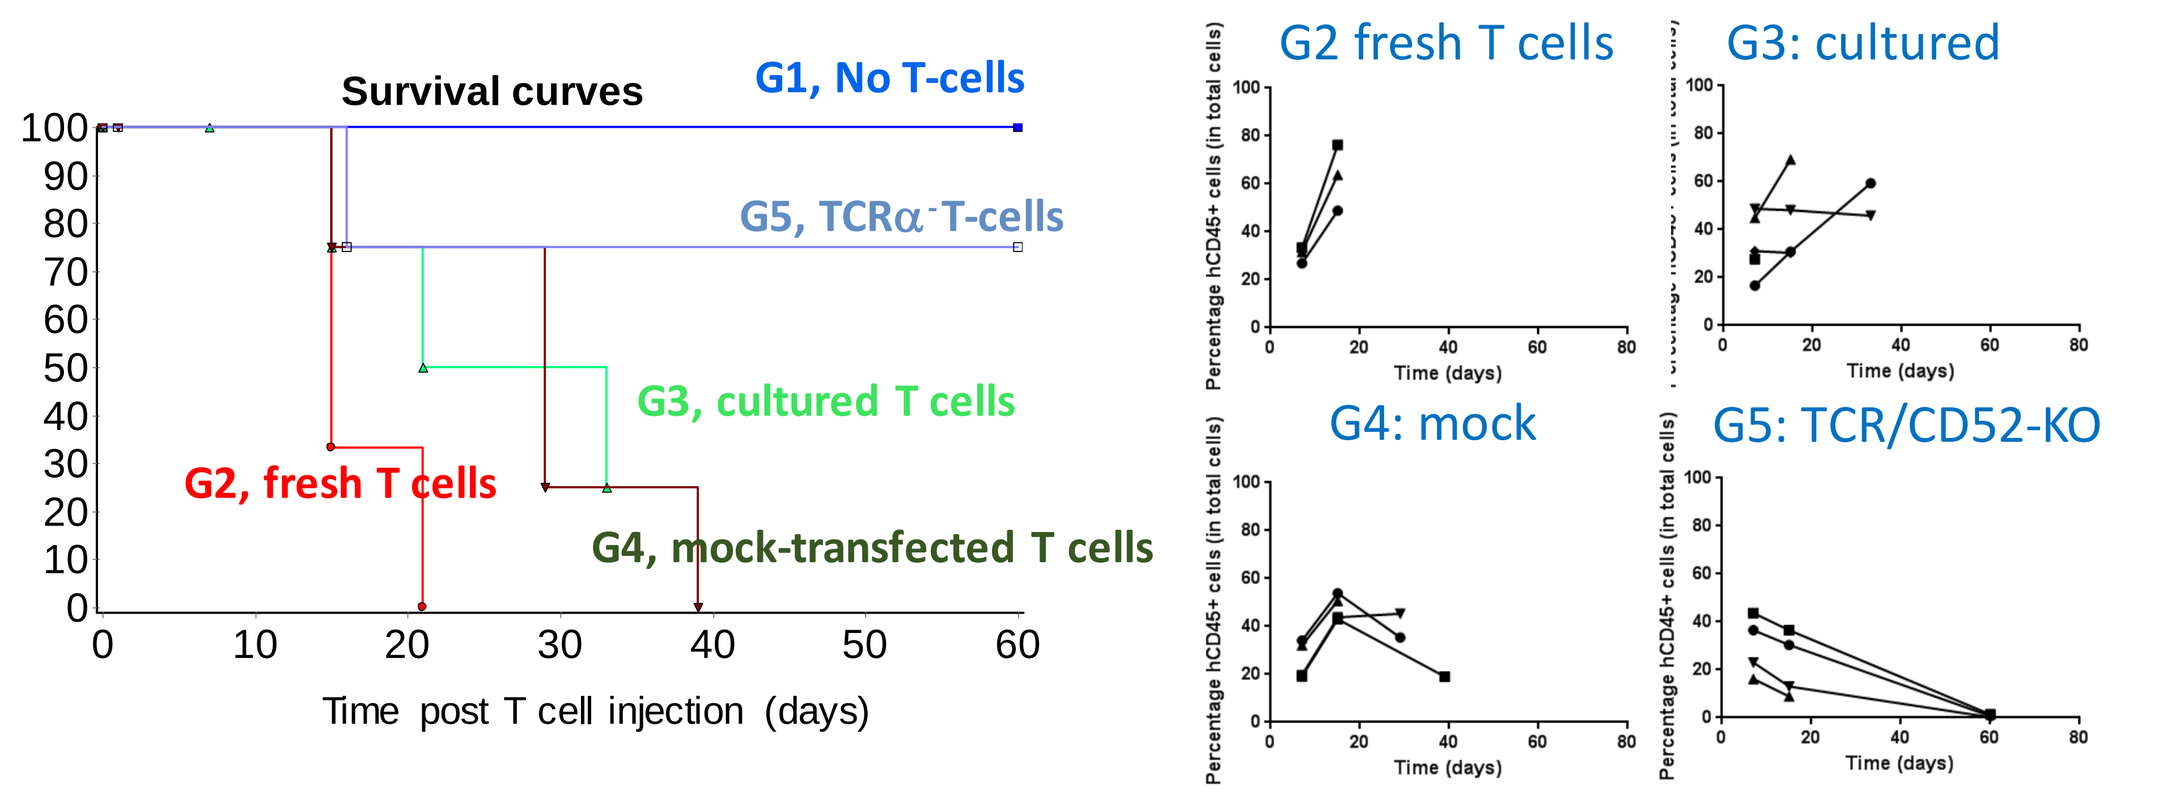
Survival curves
G1, No T-cells
G5, TCRα- T-cells
G3, cultured T cells
G2, fresh T cells
G4, mock-transfected T cells
Time post T cell injection (days)
G2 fresh T cells
G3: cultured
G4: mock
G5: TCR/CD52-KO

No mice receiving TRAC/CD52 TALEN-treated cells showed signs of GvHD whereas all mice receiving non engineered cells developped GvHD and had to be sacrificed. Human cells survived at least 2 weeks in all groups.

#6 UCART19 cells display efficient anti-tumor activity in presence of alemtuzumab

Anti-tumor activity in the presence of alemtuzumab: Alemtuzumab was injected into NSG mice one day prior to CD52$^{+/-}$ Raji-ffluc cell injection (2.5x10^5 CD52$^{+/-}$ Raji-ffluc, 30% CD52-deficient), and dKO-CART19 cells were intravenously injected the following day.

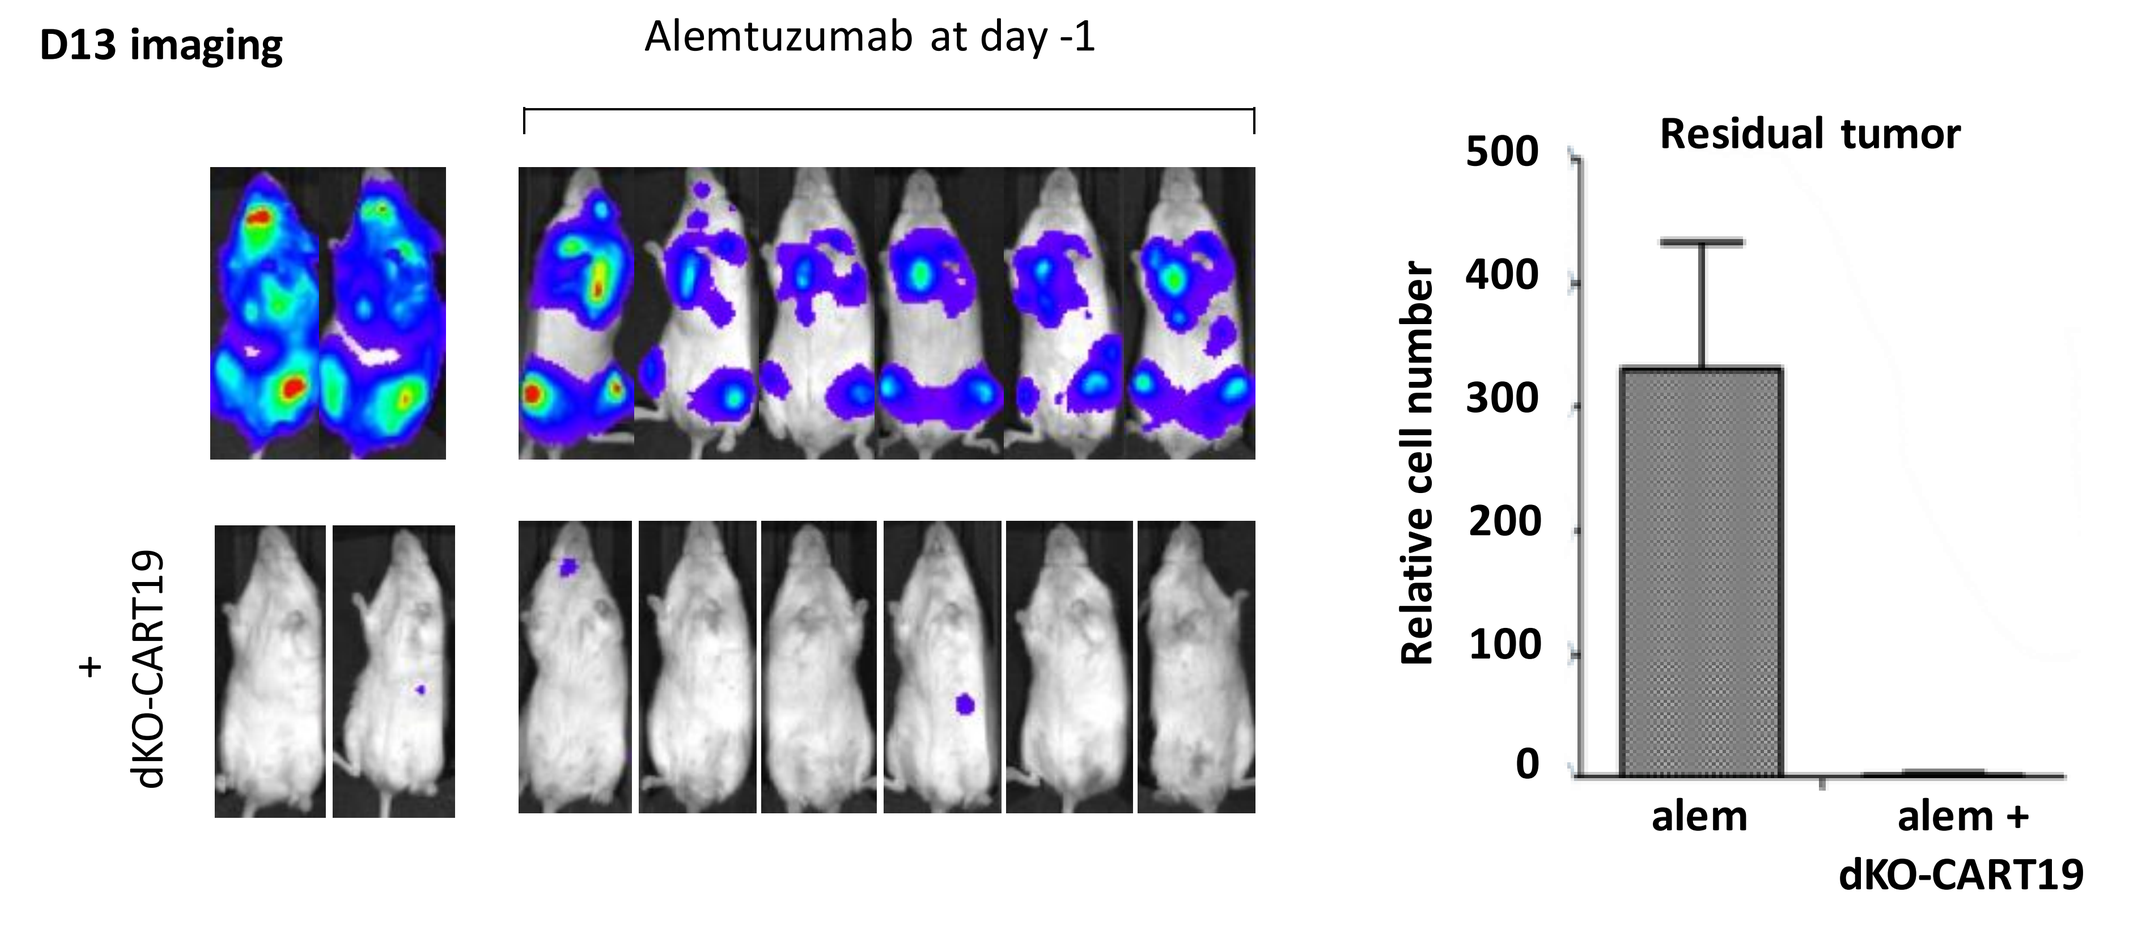
D13 imaging
Alemtuzumab at day -1
+ dKO-CART19
Residual tumor
Relative cell number
alem
alem + dKO-CART19

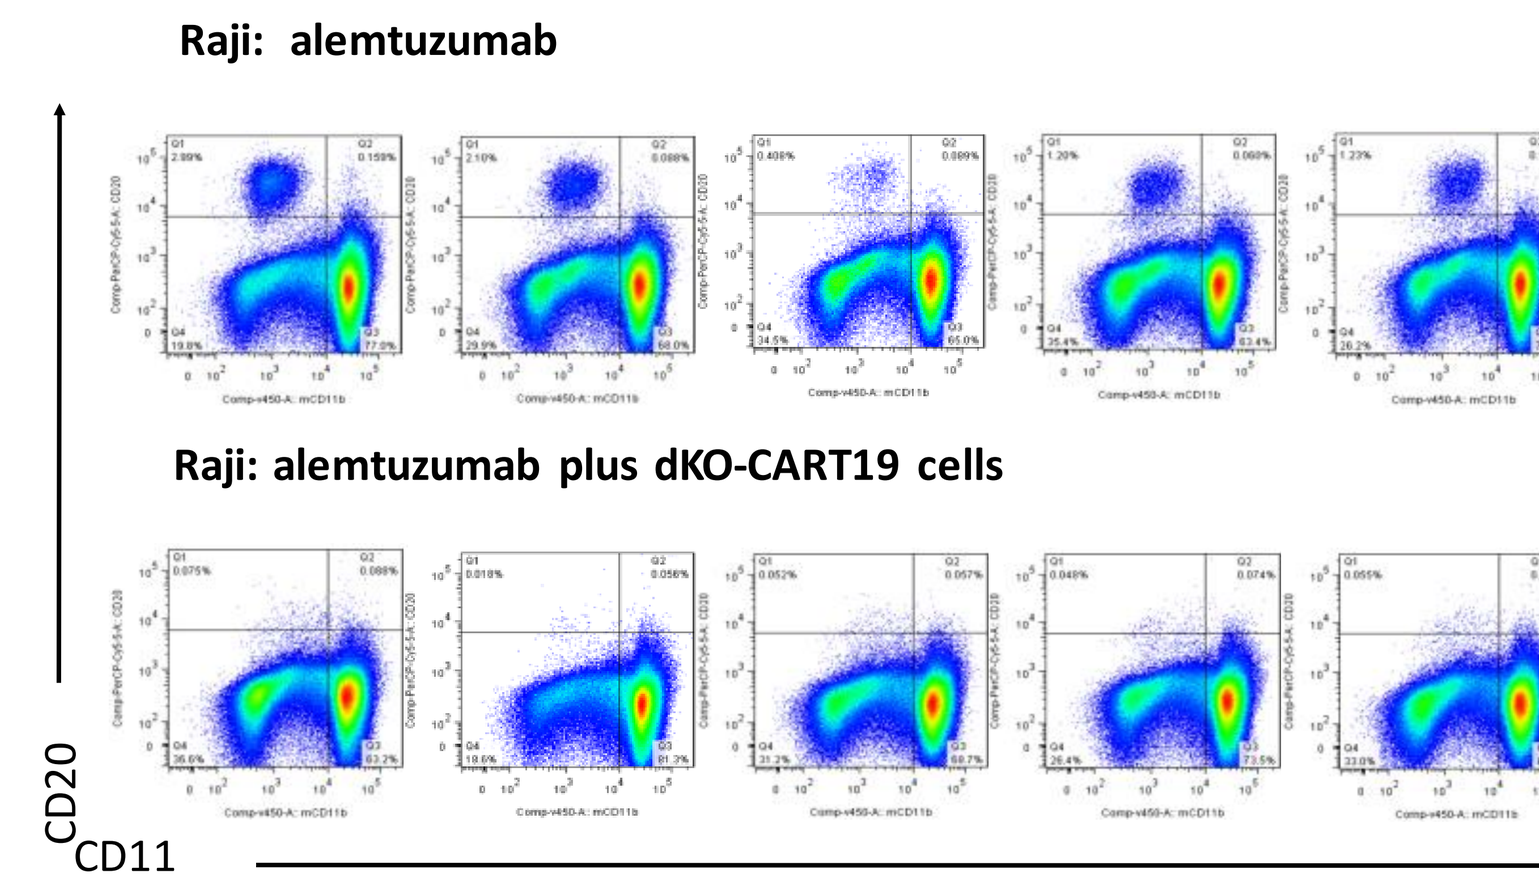
Raji: alemtuzumab
Raji: alemtuzumab plus dKO-CART19 cells
CD20
CD11

NSG mice treated with Alemtuzumab combined with UCART19 cells display complete elimination of Raji-ffluc cells from their preferred engraftment niche, bone marrow, as assessed by luminometry or flow cytometry of cell suspensions obtained from bone marrow isolated at 14 days post-injection.

#7 Absence of off target cleavage in UCART19 cells

Using recent data quantifying the impact of a substitution in a target sequence on the efficiency of TALEN cleavage we identified the top 15 potential off-target sequences in the human genome. Using deep sequencing analysis, we then examined the frequency of mutations found at these loci in UCART19 cells.

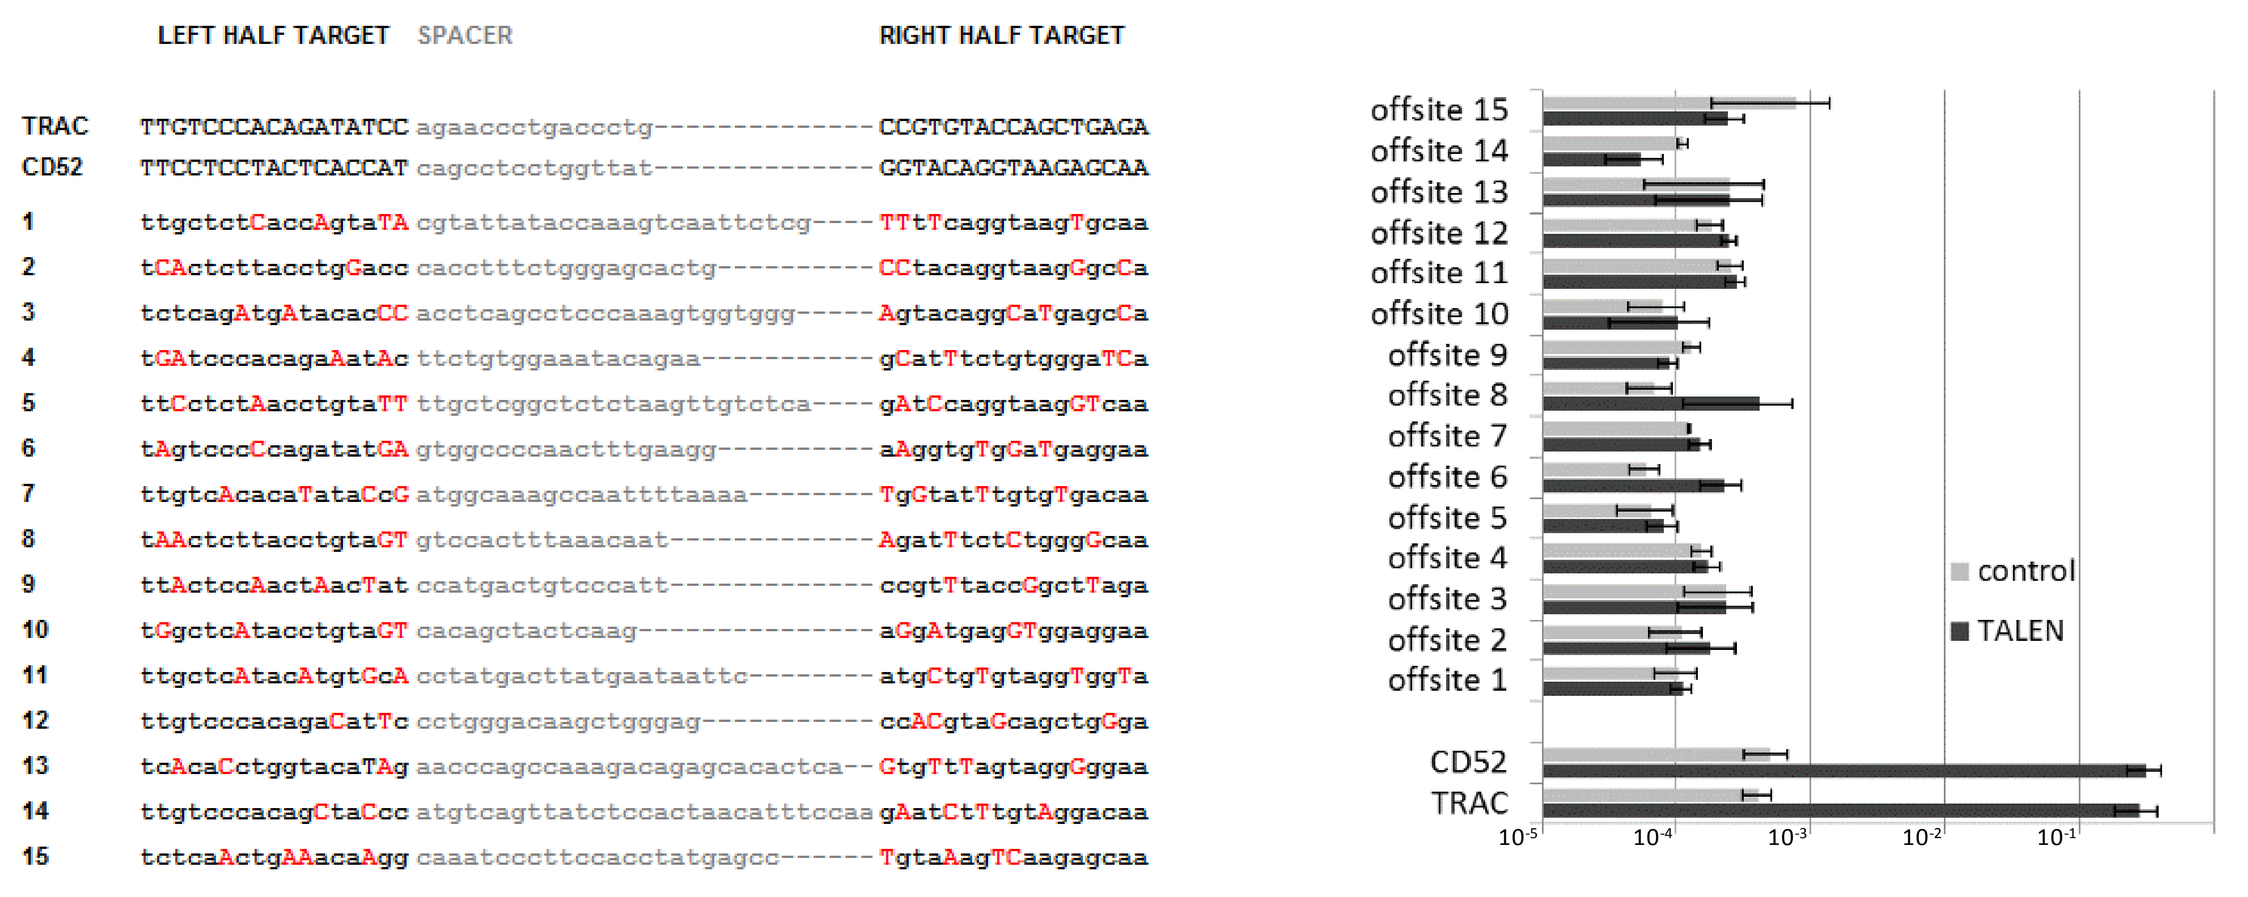
LEFT HALF TARGET
SPACER
RIGHT HALF TARGET
TRAC
CD52
control
TALEN

The highest observed frequency of insertion/deletion at any of the offsite targets was 1 in 8×10^{-4} (black bars)—a rate that is statistically indistinguishable from what we observe in control amplicons (gray bars). These results demonstrate an exceptionally low rate of offsite target cleavage—at least 600 times less likely than the intended targets.

#8 Efficient depletion of RQR8 positive cells in vivo

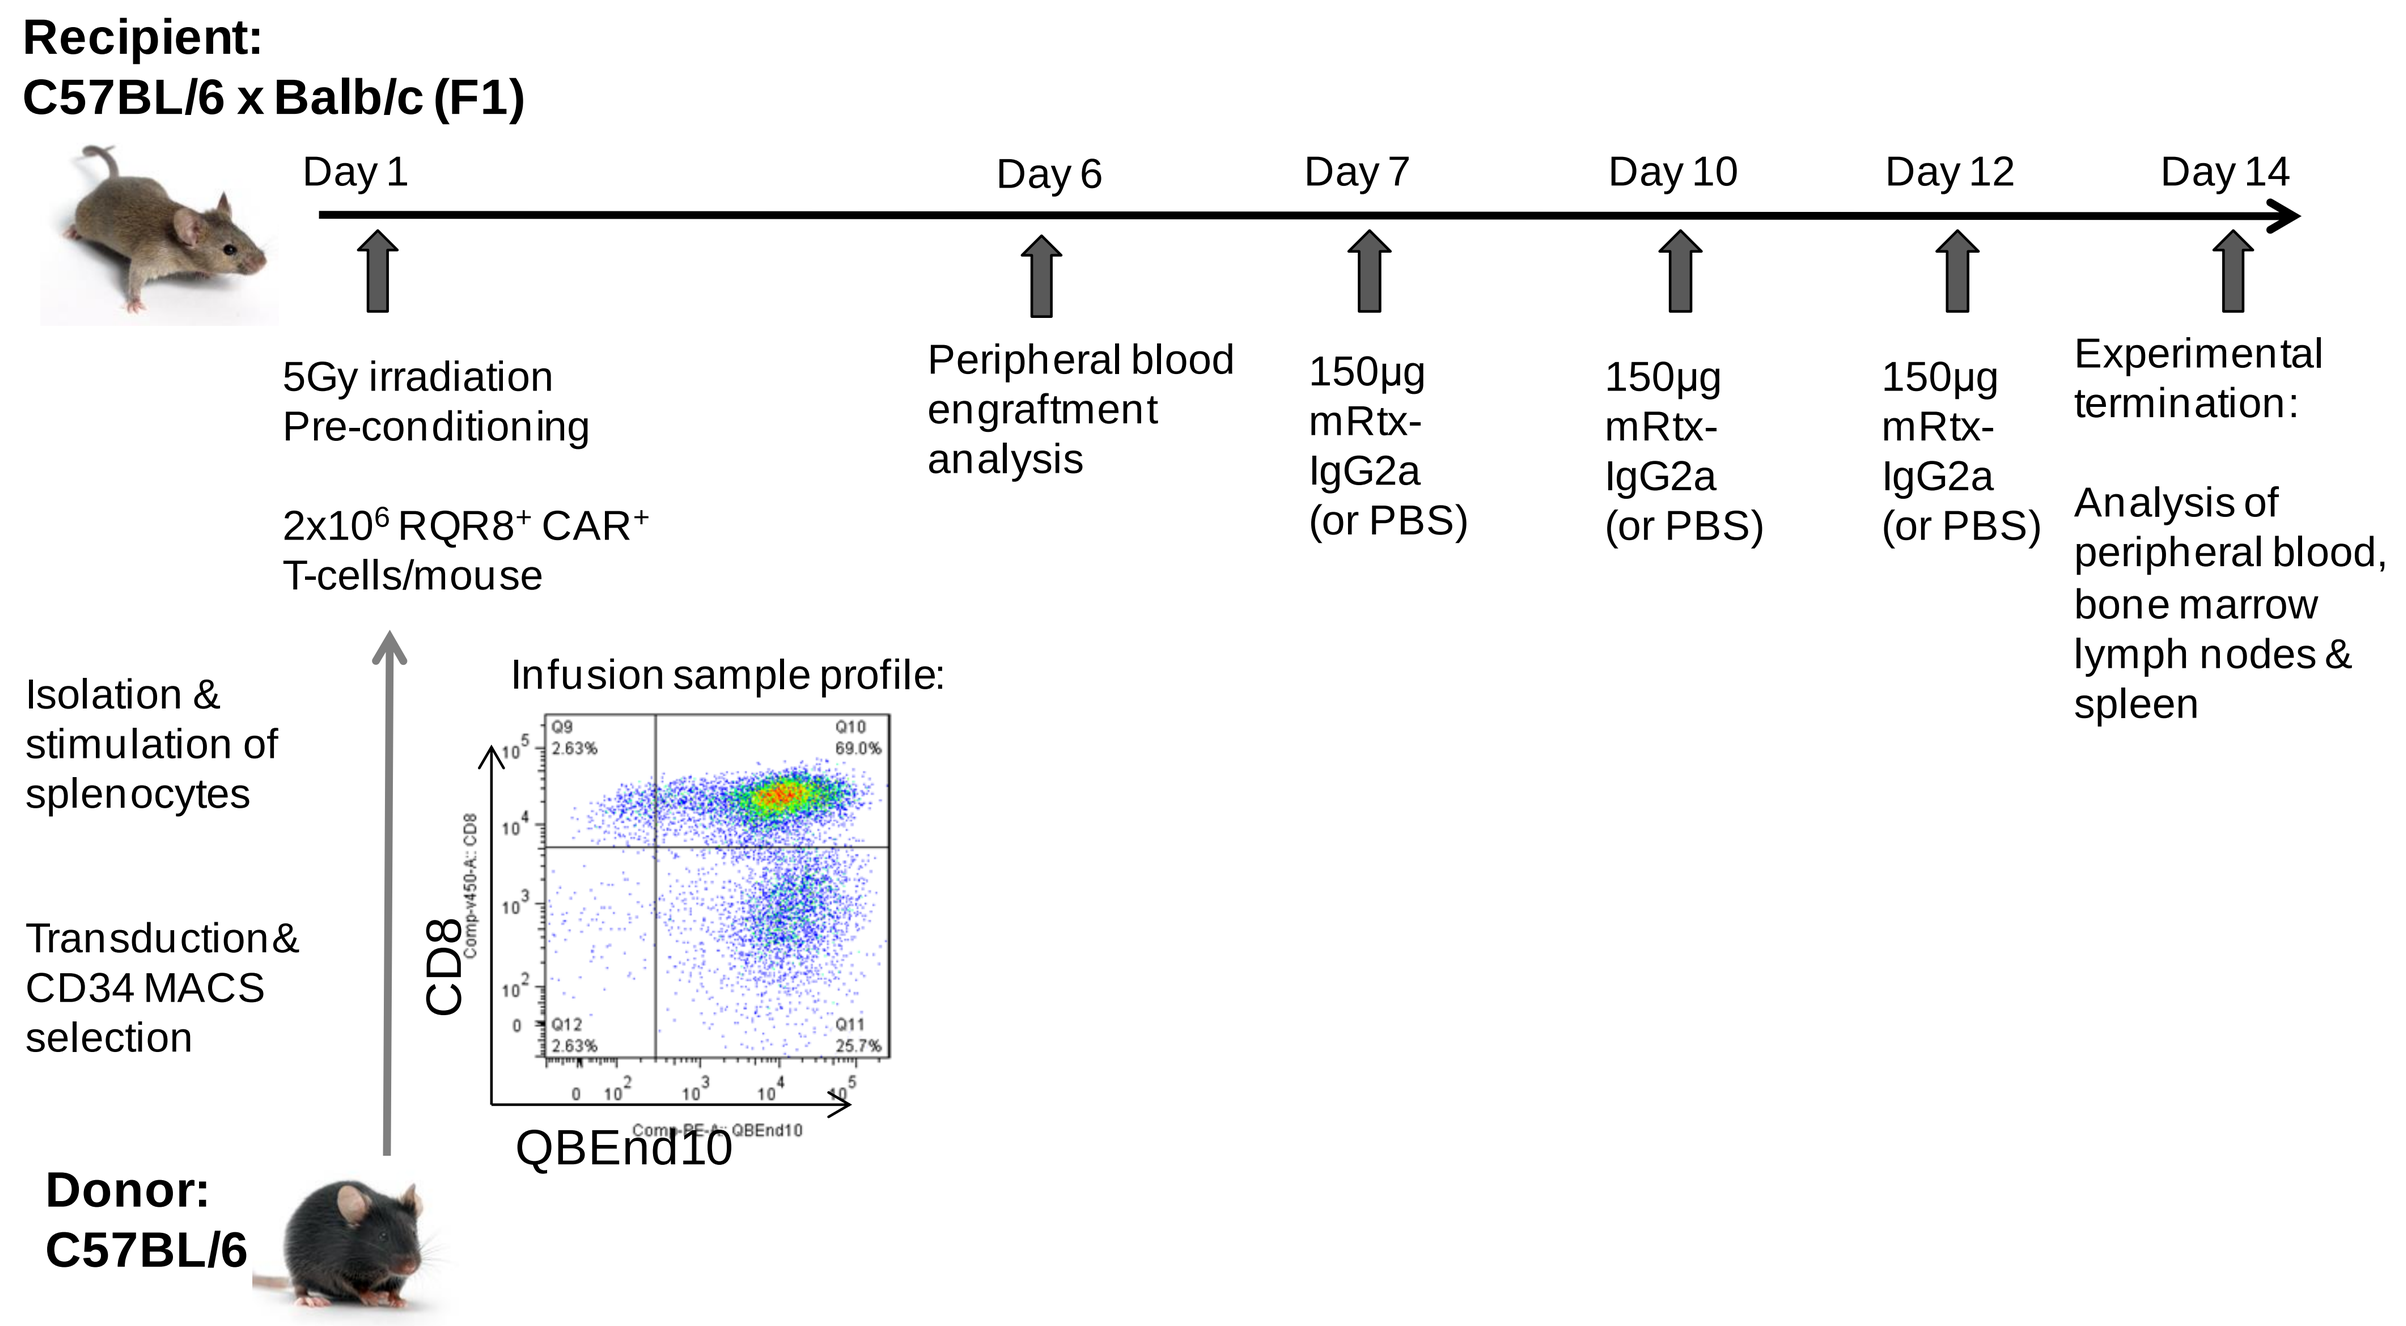
Recipient:
C57BL/6 x Balb/c (F1)
Day 1
Day 6
Day 7
Day 10
Day 12
Day 14
5Gy irradiation Pre-conditioning
2x10^6 RQR8+ CAR+ T-cells/mouse
Peripheral blood engraftment analysis
150µg mRtx-IgG2a (or PBS)
Experimental termination:
Analysis of peripheral blood, bone marrow lymph nodes & spleen
Isolation & stimulation of splenocytes
Transduction & CD34 MACS selection
Infusion sample profile:
CD8
QBEnd10
Donor:
C57BL/6

The susceptibility to Rituximab of RQR8-positive cells was shown *in vivo* in an immunocompetent mouse model using a re-engineered Rituximab to a mouse IgG2a, the functional equivalent of human IgG1 (mRtx-IgG2a) (Philip et al., 2014).

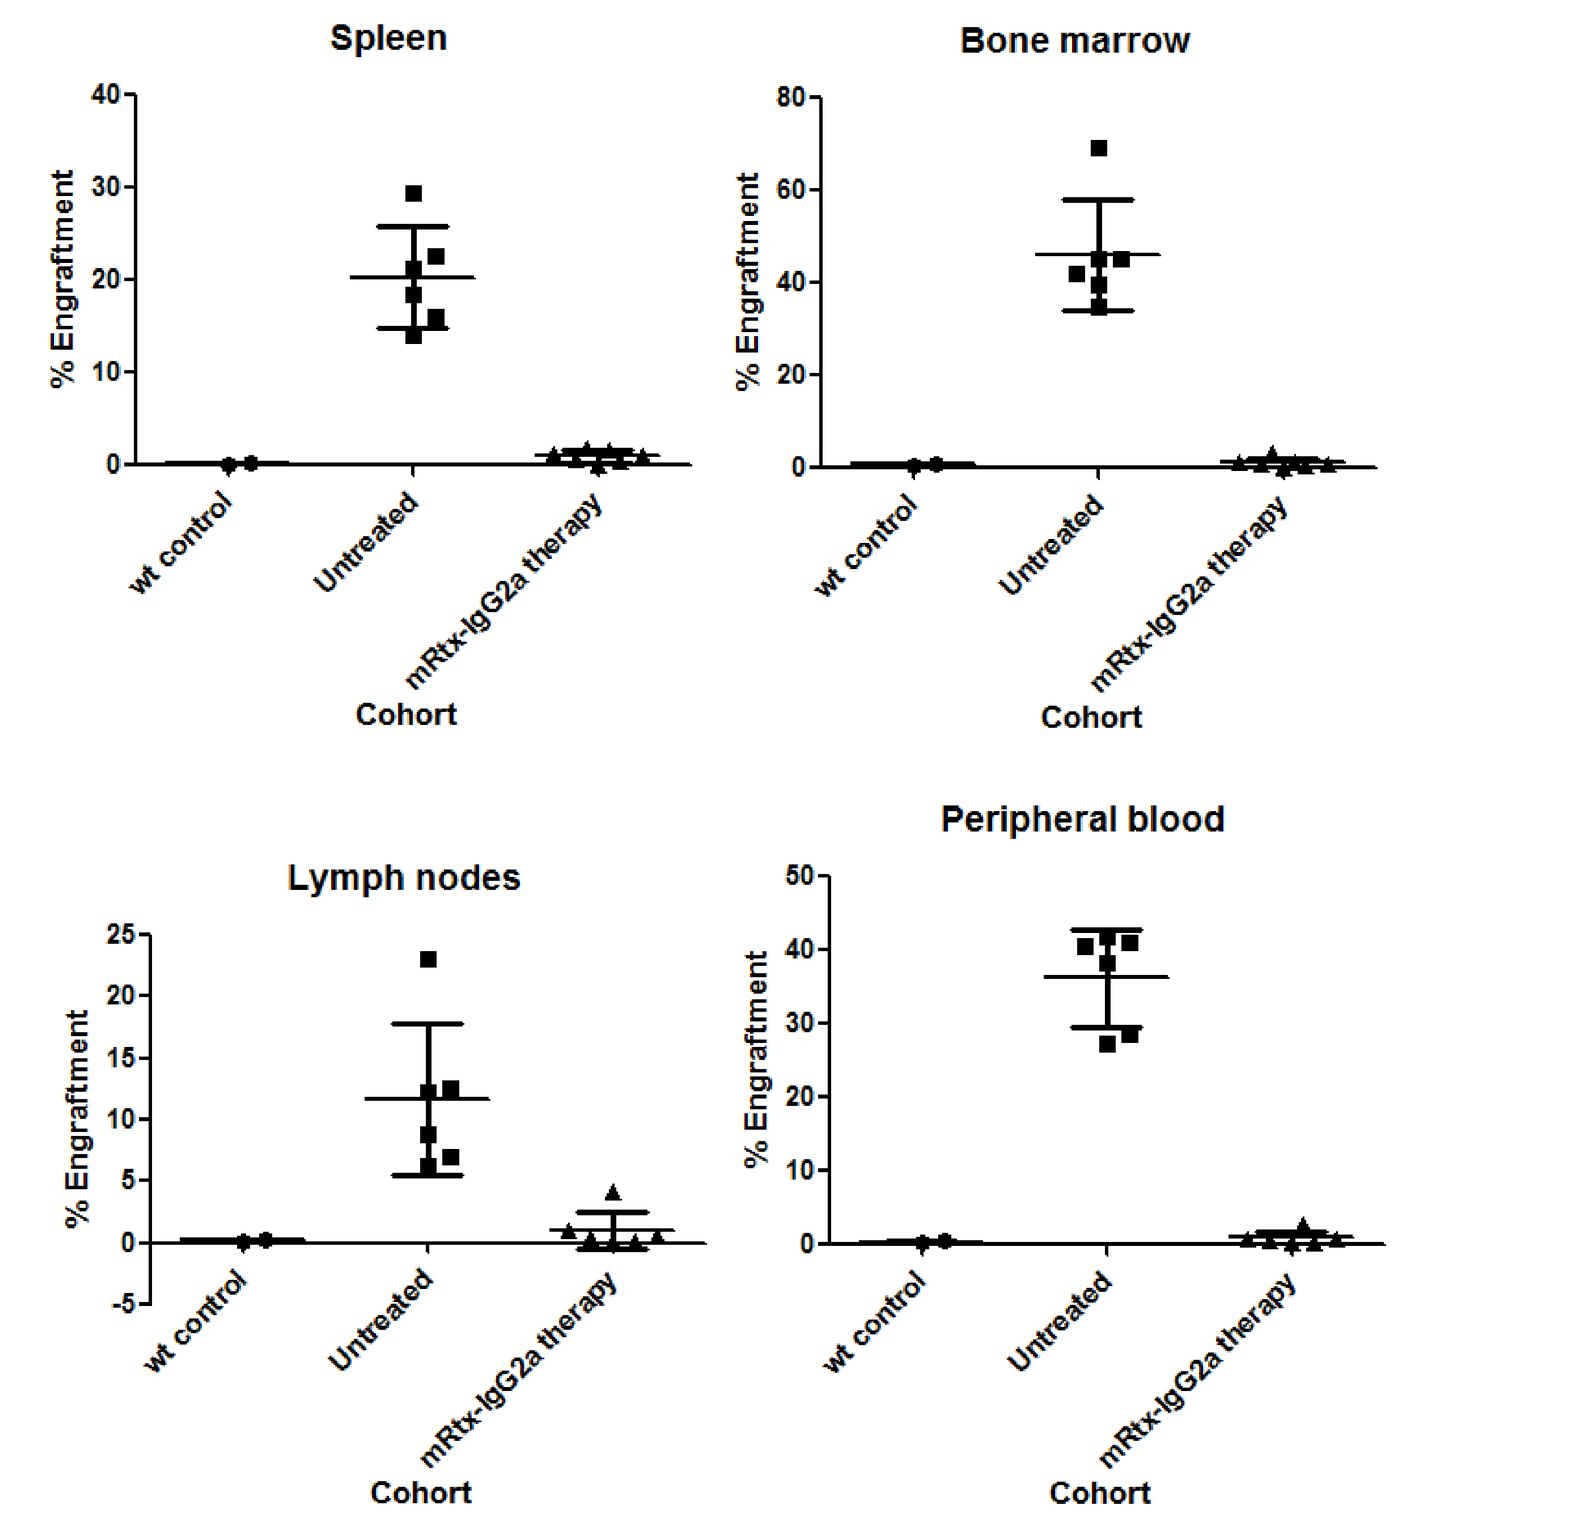
Spleen
Bone marrow
Lymph nodes
Peripheral blood
% Engraftment
wt control
Untreated
mRtx-IgG2a therapy
Cohort

Flow cytometry analysis at Day 14 of spleen, bone marrow, lymph nodes and blood indicate a significant depletion of RQR8+ cells in mice treated with Rituximab.

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis and its business. Cellectis is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise.
Cellectis proprietary information

Adoptive Immunotherapy of Multiple Myeloma with allogeneic CAR T-cells targeting CS1: enhancement of CAR activity through CS1 gene inactivation in effector cells

Roman Galetto[1], Isabelle Chion-Sotinel[1], Patricia Françon[1] and Julianne Smith[1]

[1] Cellectis SA, Paris, France

#1 Abstract

Background: Chimeric antigen receptor (CAR)-redirected T-cells have been successfully used in patients with refractory leukemia by targeting the CD19 molecule, yielding long-term durable remissions and raising hopes that a wider application of CAR technology may lead to a new paradigm in cancer treatment. Nevertheless, the CAR approach is restricted to targeting antigens expressed at the tumoral cell surface but absent from T-cells, since CAR expression will deplete antigen-expressing T-cells and thus reduce the chances to efficiently produce CAR specific T-cells.
Multiple Myeloma (MM) is a B-cell neoplasia characterized by clonal expansion of malignant plasma cells in the bone marrow, and although currently available therapies can improve overall survival it still remains incurable in most patients. Immunotherapy against MM is one area in which extensive research is being made, with novel antigenic targets being considered to drive eradication of plasma cells. Among these, the CS1 glycoprotein is highly expressed on tumor cells from most of patients with MM, making it an attractive antigen for CAR targeting.
Aims: Expression of CS1 on normal CD8+ T-cells is potentially an obstacle for the development of CAR T-cells targeting CS1. The objective of this work was therefore to evaluate if CS1 expression on T-cells could have an impact on CAR activity.
Methods: To address this limitation, TALEN® gene editing technology was used to inactivate the CS1 gene in T-cells, prior to transduction with a lentiviral vector encoding an anti-CS1 CAR.
Results: We will present data showing that non-gene-edited T-cells expressing an anti-CS1 CAR display limited cytolytic activity against MM cells expressing the CS1 antigen as well as a progressive loss of CD8+ T-cells in the effector cell population. In contrast, CS1-gene-edited CAR cells display significantly increased cytotoxic activity with the percentage of CD8+ T-cells remaining unaffected. Experiments in an orthotopic MM mouse model are currently ongoing, in order to investigate the impact of CS1 gene disruption on in vivo anti-tumoral activity.

Conclusions and Perspectives

Gene editing technology could be used to manufacture T-cells from third-party healthy donors to generate allogeneic "off-the-shelf" engineered CAR+ T-cell–based frozen products.

We have previously demonstrated that TALEN® mediated inactivation of the TCRa constant (TRAC) gene can be achieved at high frequencies and eliminate the potential for edited T-cells to mediate Graft versus Host Disease (GvHD).

We show here that TALEN®–mediated disruption of the CS1 gene in T-cells is efficient and improves in vitro activity of T-cells harboring an anti-CS1 CAR.

Multiplex genome editing can lead to the production of double KO (TRAC and CS1) T-cells, allowing large scale manufacturing of allogeneic, non alloreactive CS1 specific T-cells that could display enhanced antitumor activity.

This technology therefore offers the possibility of developing an off-the-shelf cell therapy product that would be immediately available for administration to a large number of MM patients.

#2 Allogeneic approach targeting CS1 to treat MM

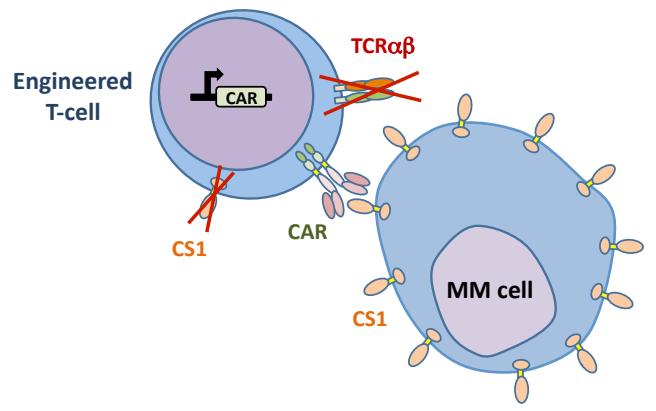

Engineered T-cell
CAR
TCRαβ
CS1
CAR
CS1
MM cell

One of the key barriers to the adoptive transfer of 3rd party CAR T-cells can be overcome via the application of TALEN® gene editing technology, deletion of the TCR gene and thus the use of any donors' T-cells without the risk of GvHD.
This technology can be used to also inactivate the CS1 gene in the same cell, allowing a more efficient production of allogenic anti-CS1 CAR T-cells.

#3 TCR alpha inactivation prevents Graft vs. Host disease

TALEN® were designed to recognize and cleave the coding sequence of the human TCRalpha gene

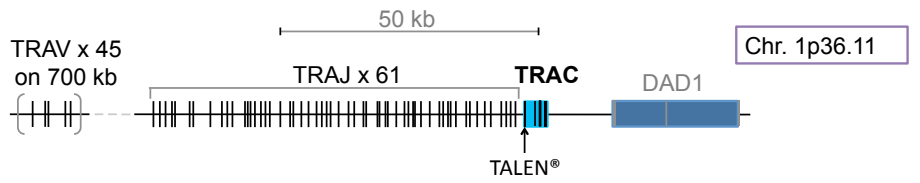

50 kb
Chr. 1p36.11
TRAV x 45
on 700 kb
TRAJ x 61
TRAC
DAD1
TALEN®

High levels of gene inactivation can be obtained in primary T-cells by electrotransfer of TALEN® mRNAs using PulseAgile electroporation technology.

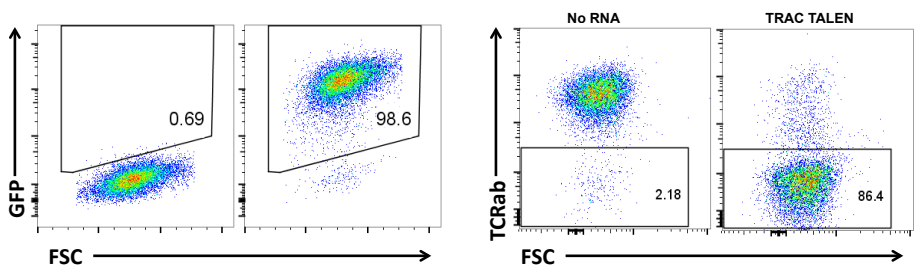

0.69
98.6
GFP
FSC
No RNA
TRAC TALEN
2.18
86.4
TCRab
FSC

Xenogeneic model of Graft vs. Host Disease: Irradiated NOG mice were injected with PBS (group 1), or 30x10⁶ human T-cells. The cells were obtained from activated PBMCs after 3 day expansion (group 2), 15-day expansion (group 3), mock electroporation and 15-day expansion (group 4), or TRAC TALEN®-electroporation, 15-day expansion and depletion of TCR+ cells (group 5).

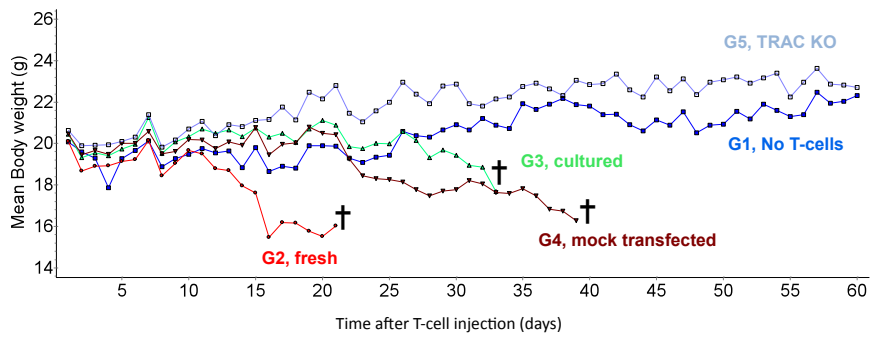

Mean Body weight (g)
G5, TRAC KO
G1, No T-cells
G3, cultured
G2, fresh
G4, mock transfected
Time after T-cell injection (days)

No mice receiving TRAC TALEN®-treated cells showed signs of GvHD, whereas all mice receiving non engineered cells developed GvHD and had to be sacrificed. Human cells survived at least 2 weeks in all groups.

#4 Generation of an anti-CS1 Chimeric Antigen Receptor

An scFv targeting the extracellular domain of the human CS1 antigen (SLAMF7) was used to construct a chimeric antigen receptor containing a 41BB co-stimulatory domain and the CD3z activation domain.
A lentiviral vector was constructed in which CAR expression was driven by an hEF1a promoter and coupled to the Blue Fluorescent Protein through a 2A ribosomal skip peptide.

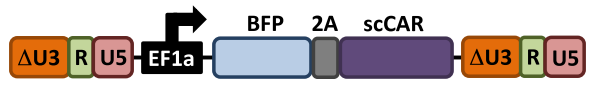

ΔU3
R
U5
EF1a
BFP
2A
scCAR
ΔU3
R
U5

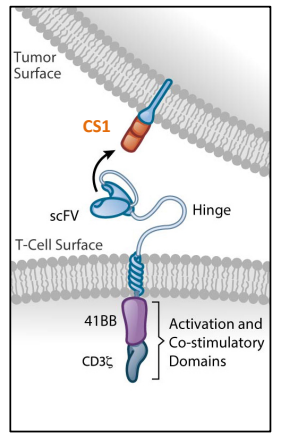

Tumor Surface
CS1
scFV
Hinge
T-Cell Surface
41BB
CD3ζ
Activation and Co-stimulatory Domains

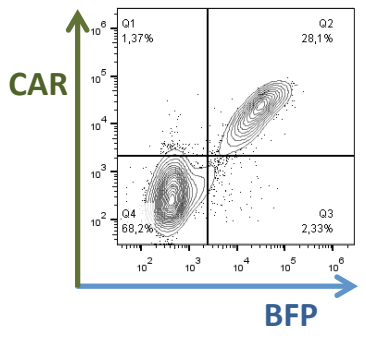

CAR
BFP
Q1 1,37%
Q2 28,1%
Q4 68,2%
Q3 2,33%

CAR expression in human T-cells was assessed by flow cytometry, using a recombinant protein containing the EC domain of the CS1 protein fused to a mouse Fc fragment. Binding of this protein to CAR+ cells was detected using a fluorescent anti-Fc antibody.
As shown in the flow data, CAR expression correlates with the BFP signal of transduced cells.

#5 CS1 gene inactivation in human T-cells

TALEN® targeting the coding sequence of the human CS1 gene were designed, and the corresponding mRNAs were transfected in primary T-cells using PulseAgile electroporation technology.
Flow cytometry analysis revealed that CS1 is predominantly expressed in CD8+ T-cells, and high levels of CS1 gene inactivation were obtained in this cellular population.

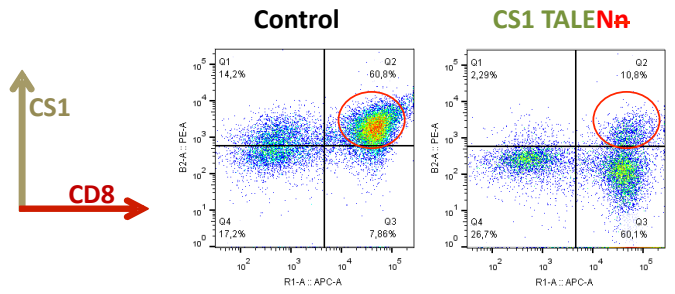

Control
CS1 TALEN~~n~~
CS1
CD8
Q1 14,2%
Q2 60,8%
Q4 17,2%
Q3 7,86%
Q1 2,29%
Q2 10,8%
Q4 26,7%
Q3 60,1%

#6 CS1 CAR expression in human T-cells

T-cells were transfected with mRNAs encoding the TALEN® targeting the CS1 gene (or mock transfected) 3 days after T-cell activation. 48h after transfection the cells were transduced with the lentiviral vector encoding the anti-CS1 CAR and amplified for 7 additional days.
At the end of this culture period T-cells were analyzed for CD8 and CAR expression by flow cytometry. 5 different T-cell donors were analyzed.

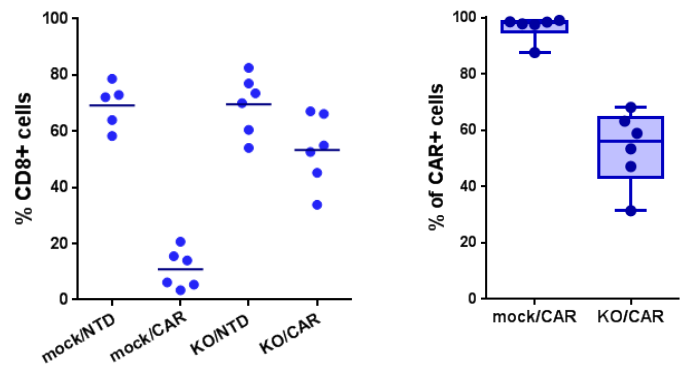

% CD8+ cells
mock/NTD
mock/CAR
KO/NTD
KO/CAR
% of CAR+ cells
mock/CAR
KO/CAR

The percentage of CD8+ cells is dramatically reduced in non-edited (mock transfected) cells upon anti-CS1 CAR expression, indicating that CD8+ cells are being targeted by CAR+ cells during the expansion step. Furthermore, a concomittant enrichment of CAR+ cells is observed in mock transfected cells compared to CS1 gene-edited cells.

#7 anti-CS1 CAR activity is enhanced in CS1 KO T-cells

Degranulation activity was assessed 8 days after transduction. CAR+ T-cells were co-cultured with CS1 expressing cells (NCI-H929 or L363) and with $CS1_{NEG}$ cells as a control.
Degranulation activity was measured by detecting CD107a expression in CD8+/CAR+ cells after 6h of co-culture. 5 different T-cell donors were analyzed.
CS1 KO cells are able to degranulate in the presence of the CS1 antigen at the same levels that non-edited cells expressing the same CAR.

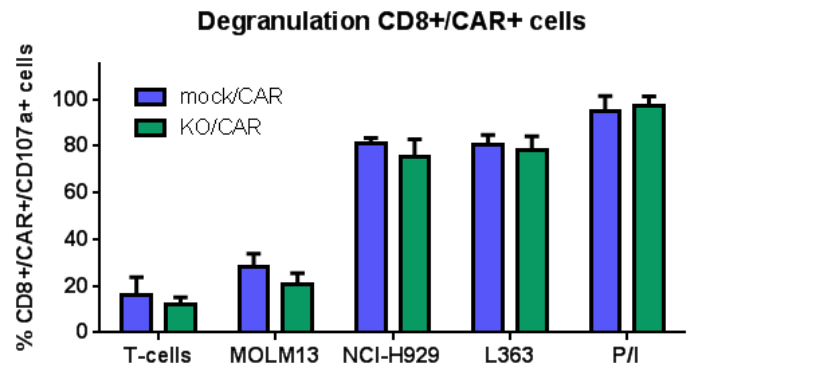

Degranulation CD8+/CAR+ cells
% CD8+/CAR+/CD107a+ cells
mock/CAR
KO/CAR
T-cells
MOLM13
NCI-H929
L363
P/I

The Cytotoxic activity of the different T-cell groups against L363 and NCI-H929 cells was also measured by flow cytometry, upon co-culturing the effector and target cells for 4h.
Activity is normalized to consider an equal transduction efficiency between mock and CS1 KO T-cells.
Even if differential activities are observed among the 5 donors tested, CS1 KO cells show a higher cytotoxic activity when compared to mock transfected cells.

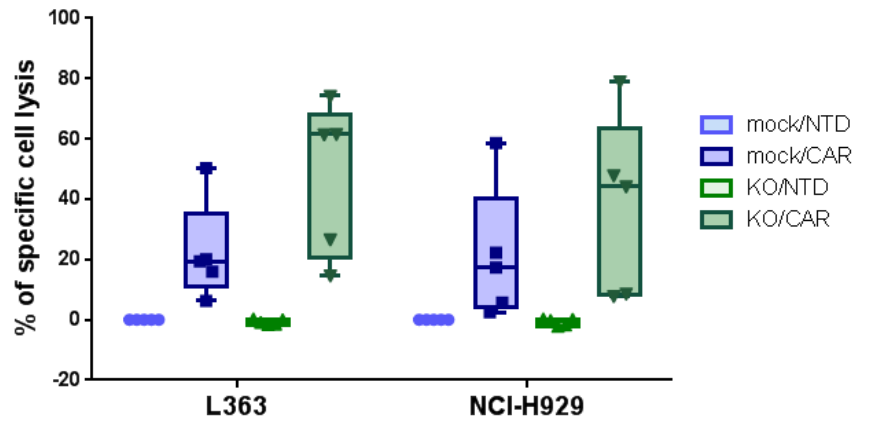

% of specific cell lysis
mock/NTD
mock/CAR
KO/NTD
KO/CAR
L363
NCI-H929

#8 UCART-CS1 anti-tumor activity in vivo

NOG mice were sublethally irradiated (1,44 Gy) 8 days before injection of T-cells. At Day (-7) 10⁶ L363-Luciferase cells/mice were iv injected.
Mice were then infused with CS1 KO T-cells, non engineered CAR+ T-cells, or UCART-CS1 cells (CS1/TCRa KO, CAR+ T-cells). Bioluminiscent signal was assessed at D(-1), D7 and D14 post injection of T-cells

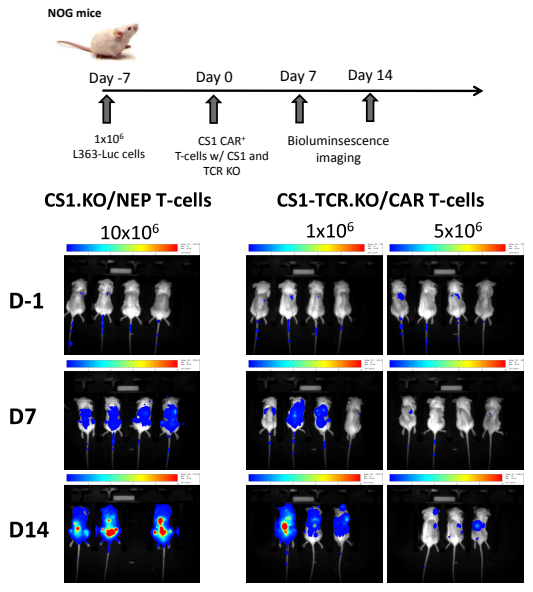

NOG mice
Day -7
Day 0
Day 7
Day 14
1x10⁶ L363-Luc cells
CS1 CAR+ T-cells w/ CS1 and TCR KO
Bioluminsescence imaging
CS1.KO/NEP T-cells
CS1-TCR.KO/CAR T-cells
10x10⁶
1x10⁶
5x10⁶
D-1
D7
D14

UCART-CS1 cells display anti-tumor activity in vivo

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis SA and its business. Cellectis SA is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. This communication contains Cellectis' proprietary information.
Talen® and Cellectis® are trademarks owned by the Cellectis Group.